L-DOS47 – a tumour microenvironment alkalization agent and a novel lung adenocarcinoma specific therapeutic candidate

Heman Chao[1],Wah Wong[1], Baomin Tian[1], Carl De Luca[1], Jianbing Zhang[2], Roger MacKenzie[2], Elda Bravo Grimaldo[3], Crystal Fulton[3], Mike Jackson[3], Axel Wellmann[4], Donald Segal[1]

1. Helix BioPharma Corp., Aurora Ontario Canada 2. NRC Institute for Biological Sciences,Ottawa Ontario Canada 3. NRC Institute for Biodiagnostics,Winnipeg Manitoba Canada 4. University Hospital Aachen, RWTH Aachen Germany

Summary

It has been observed that human solid tumours produce an acidic local microenvironment. This phenomenon can impact the efficacy of certain chemotherapeutics by altering their partitioning coefficient between extracellular and intracellular compartments due to charge alternations. This metabolic condition can also induce metastasis and confer a growth advantage to certain cancers. An immuno-conjugate cancer therapeutic has been developed to exploit the acidic tumour extracellular environment. The molecule, L-DOS47, is composed of a lung adenocarcinoma specific single domain antibody and a urease enzyme. The antibody serves as a targeting agent to deliver the enzyme to the affected sites while the urease enzyme converts urea, an abundant metabolite, into ammonia and generates a local pH increase. The combined effect of ammonia toxicity and pH increase is cytotoxic to cancer cells in culture and in xenograft models. L-DOS47 binds specifically to human lung adenocarcinoma as shown by tissue-specific staining. Imaging studies using A549 xenografts and intravenously-given labelled L-DOS47 showed that the drug molecule preferentially accumulated and persisted at the tumour site well over 72 hours, consistent with a strong binding antibody. *In vitro* experiments showed that L-DOS47 potentiated the cytotoxicity of a number of chemo-therapeutics. In some instances, less than one tenth of the normal drug dose was required to achieve the desired cytotoxic effect. In summary, L-DOS47 is a novel immuno-conjugate that preferentially targets lung tumours and generates cytotoxicity directly through its enzymatic function or in combination with weakly basic chemotherapeutics through a potentiated pH effect.

Outline

1. Proposed mechanism of action
2. L-DOS47 effect on tumour cells
3. L-DOS47 binding on human normal and cancer tissue
4. L-DOS47 enhances the effect of weakly basic drugs
5. L-DOS47 fluorescent imaging study
6. Cell line selection for xenograft study

Mechanism of Action

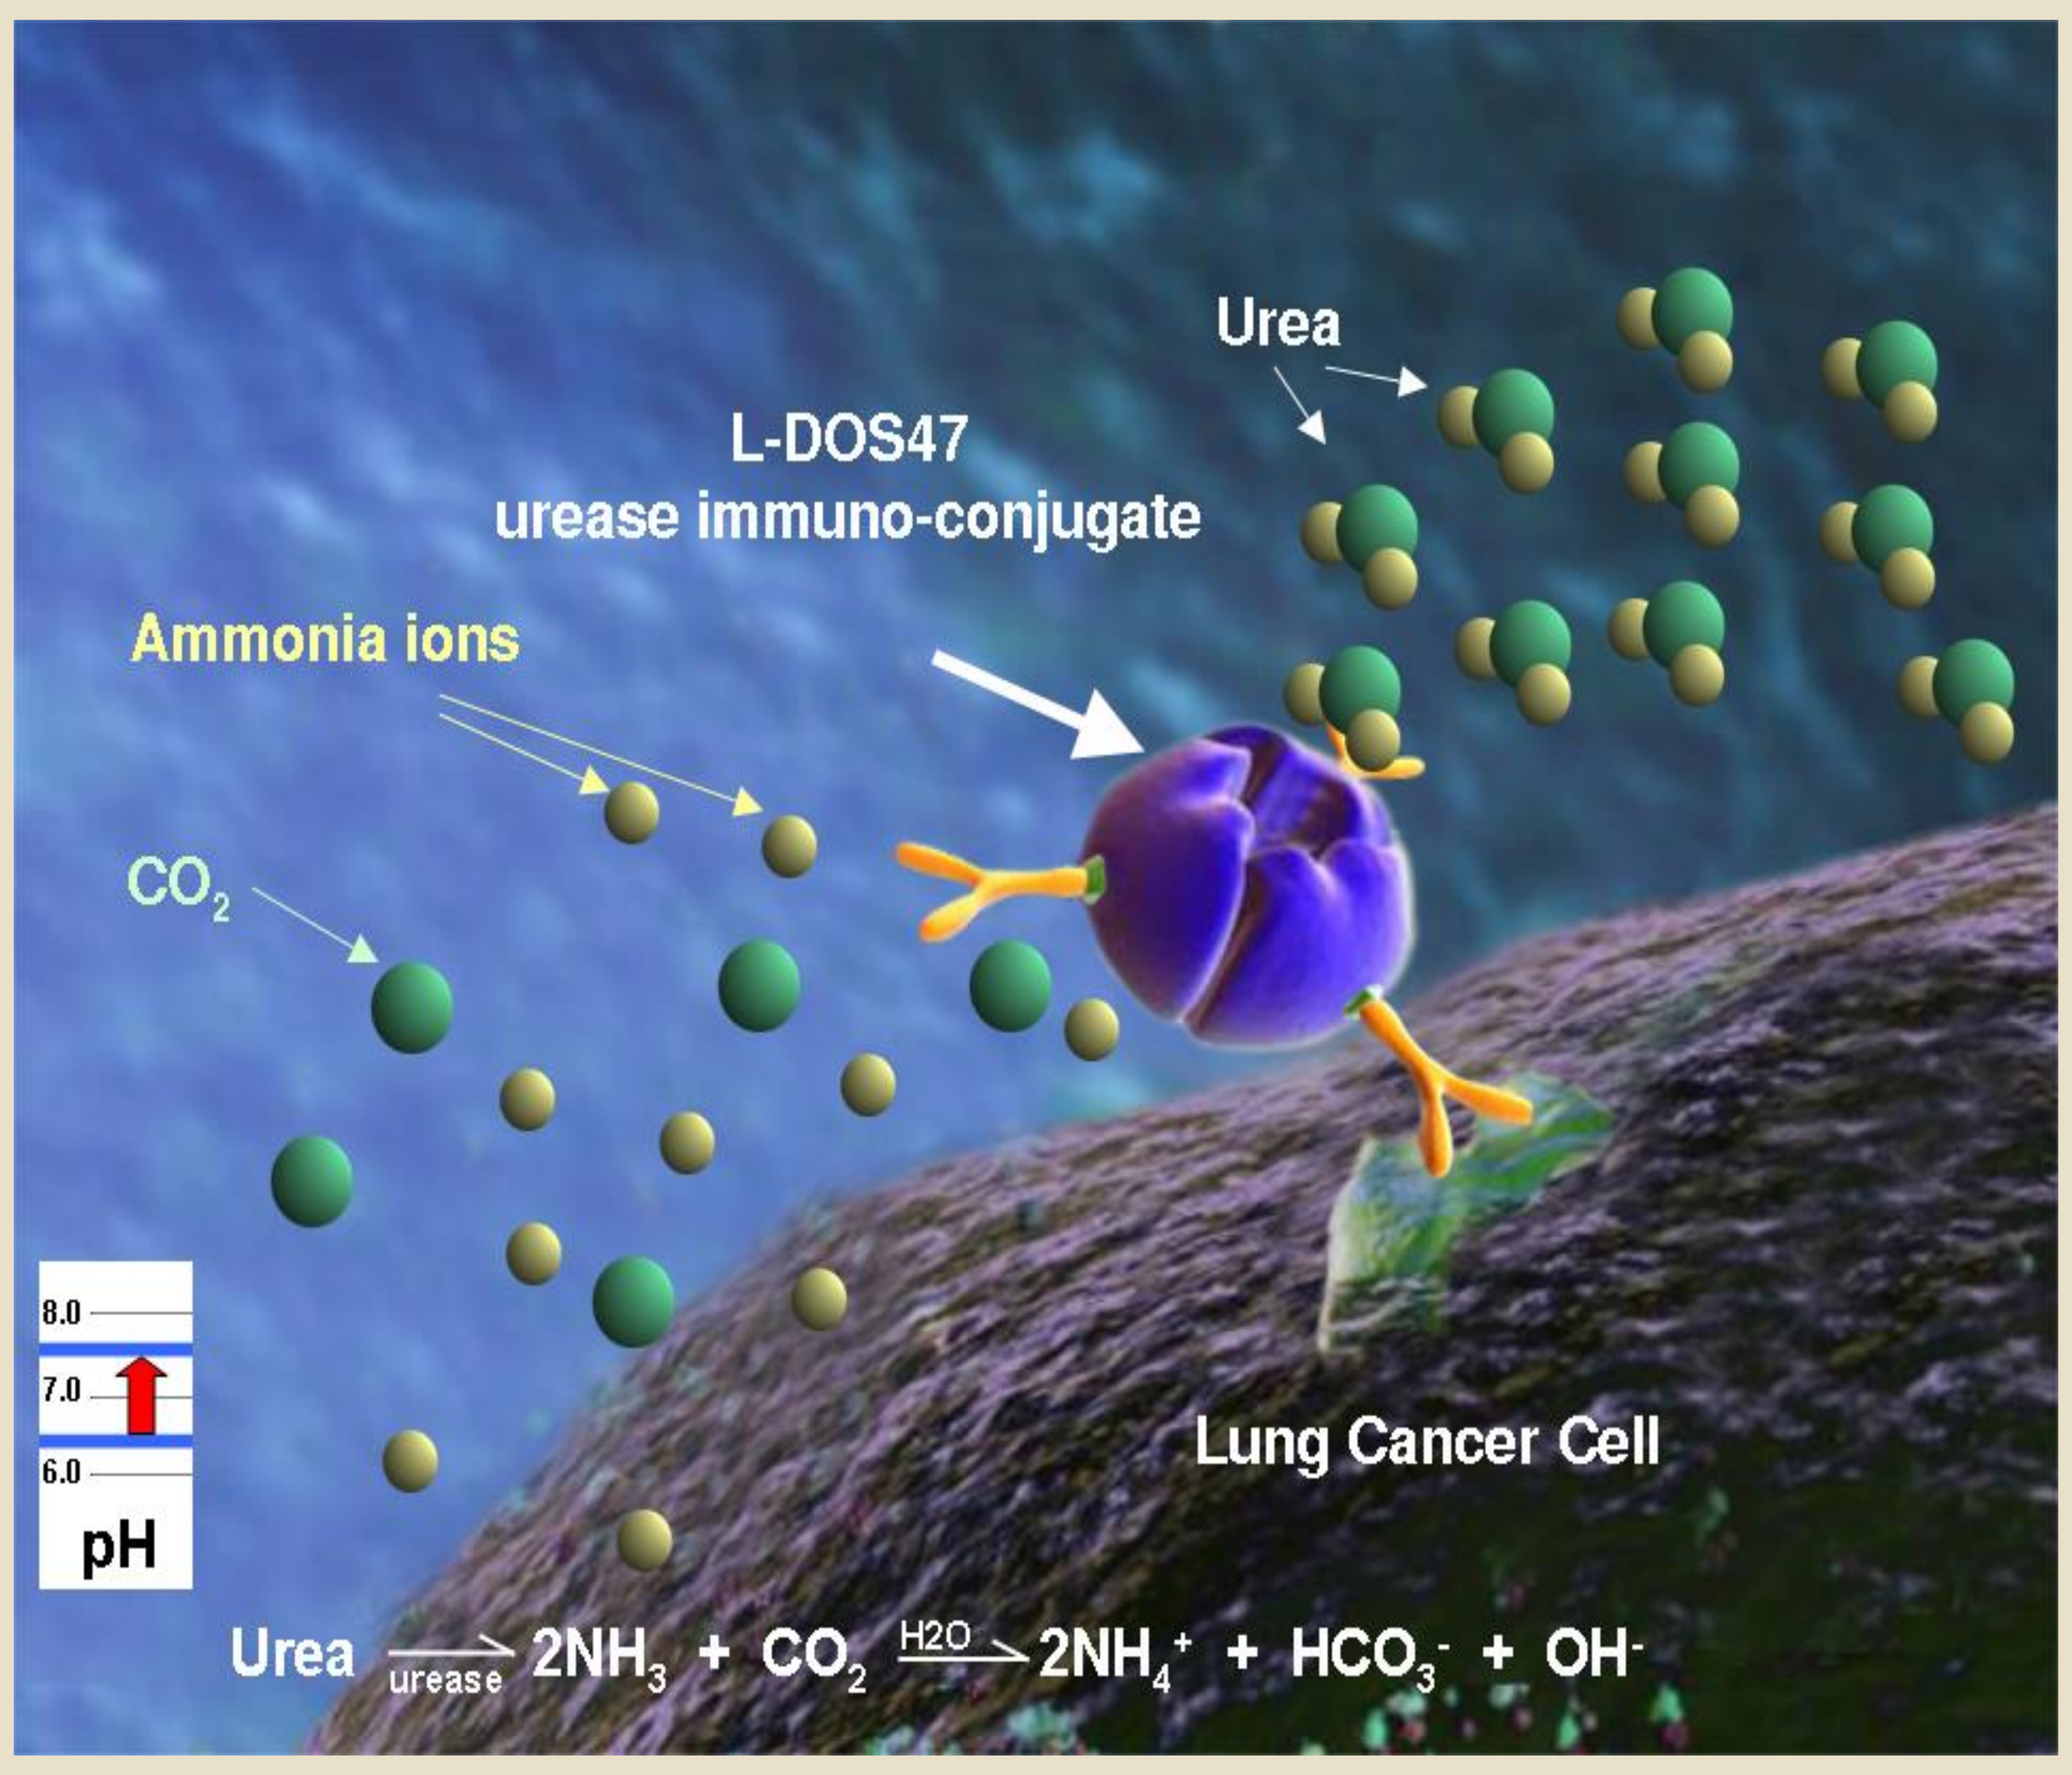
Urea
L-DOS47
urease immuno-conjugate
Ammonia ions
CO_2
8.0
7.0
6.0
pH
Lung Cancer Cell
$\text{Urea} \xrightarrow[\text{urease}]{} 2NH_3 + CO_2 \xrightarrow{H2O} 2NH_4^+ + HCO_3^- + OH^-$

L-DOS47 effect on tumour cells

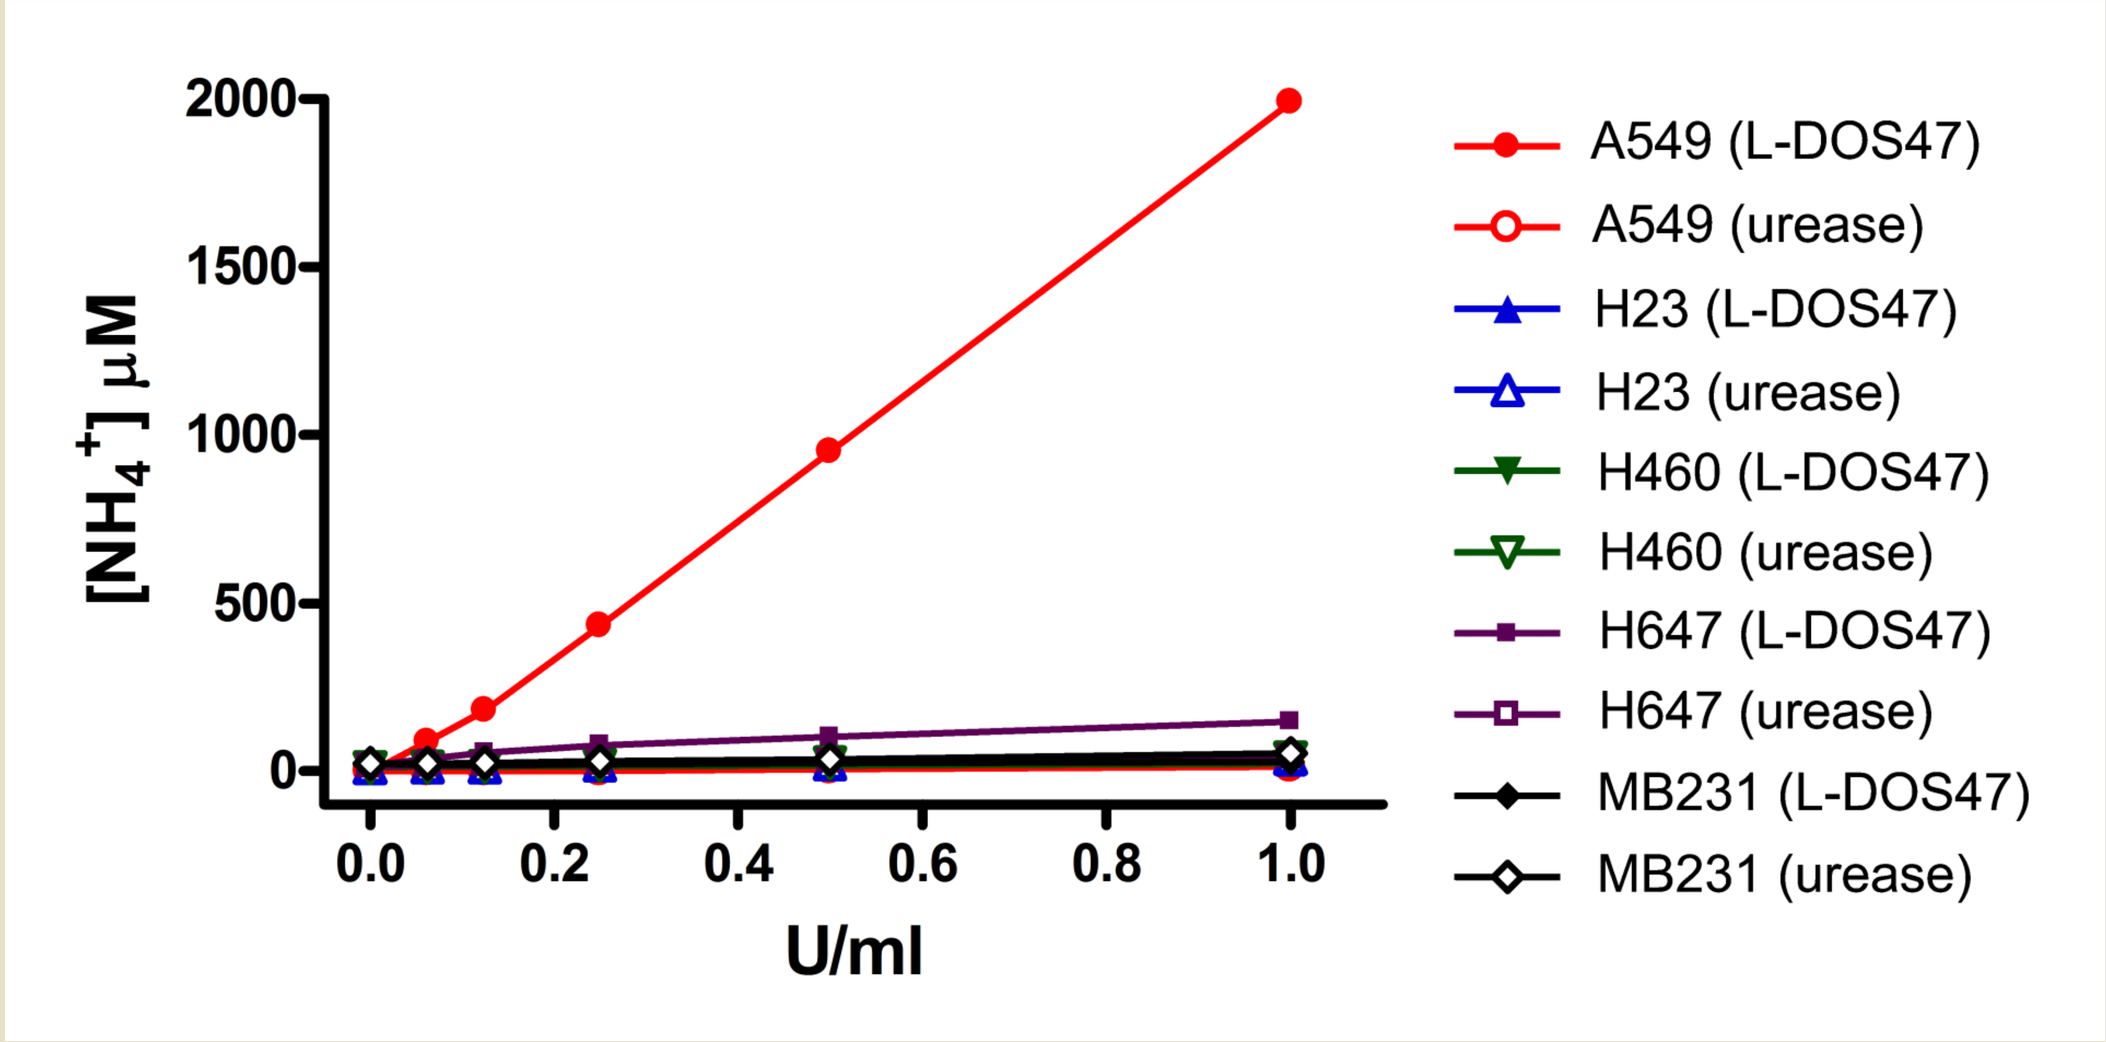
$[NH_4^+]$ µM
U/ml
A549 (L-DOS47)
A549 (urease)
H23 (L-DOS47)
H23 (urease)
H460 (L-DOS47)
H460 (urease)
H647 (L-DOS47)
H647 (urease)
MB231 (L-DOS47)
MB231 (urease)

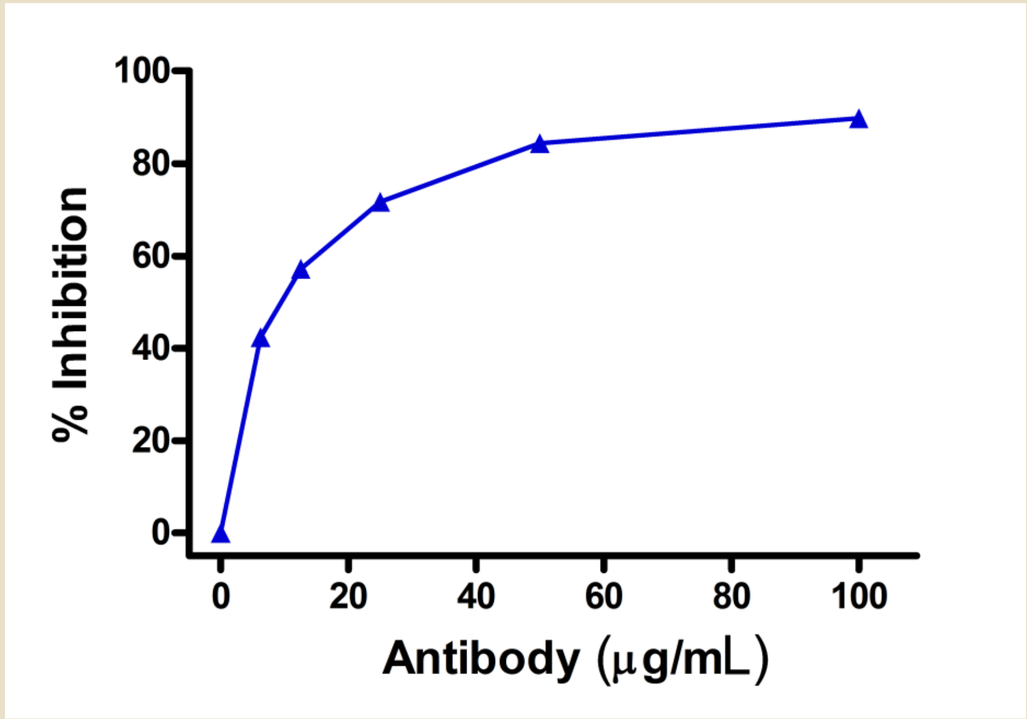
% Inhibition
Antibody (µg/mL)

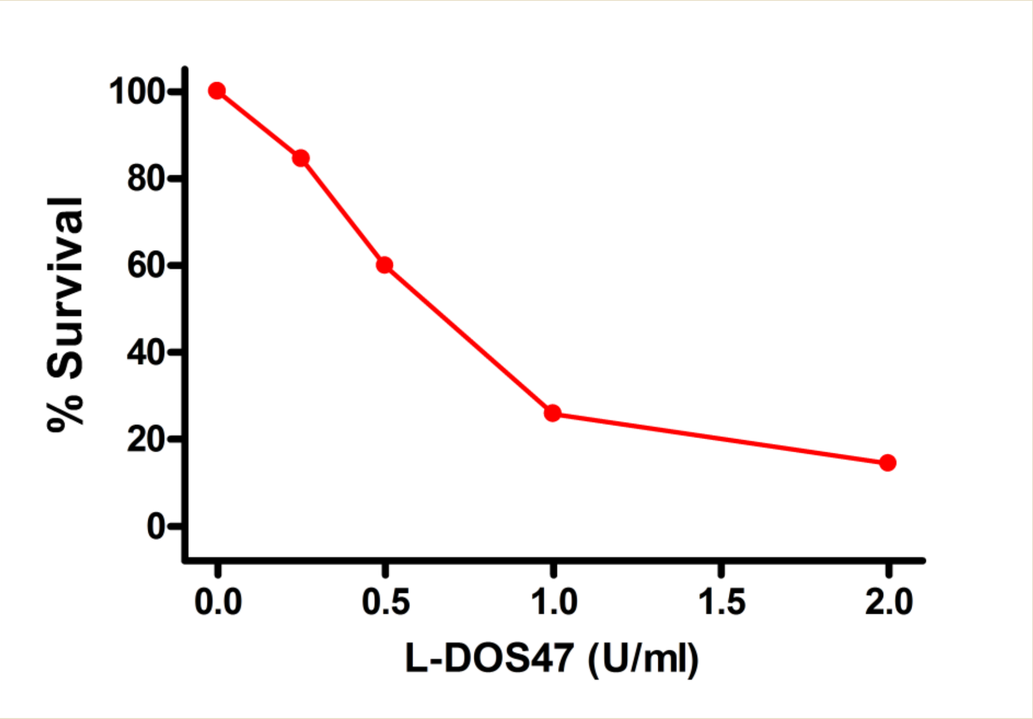
% Survival
L-DOS47 (U/ml)

Five cell lines (A549, H23, H460, H647, and MDA-MB231) were seeded in 96-well culture plates to test for the binding specificity of L-DOS47 (top). L-DOS47 binds specifically and significantly only to A549 cells – a human lung adenocarcinoma line. The immuno-conjugate also generates ammonia in the presence of urea. Specificity of the binding is confirmed by competitive displacement with the antibody alone in a dose dependent manner (bottom left). L-DOS47 is able to kill A549 cells in a dose dependent manner (bottom right).

L-DOS47 effect on tumour cells

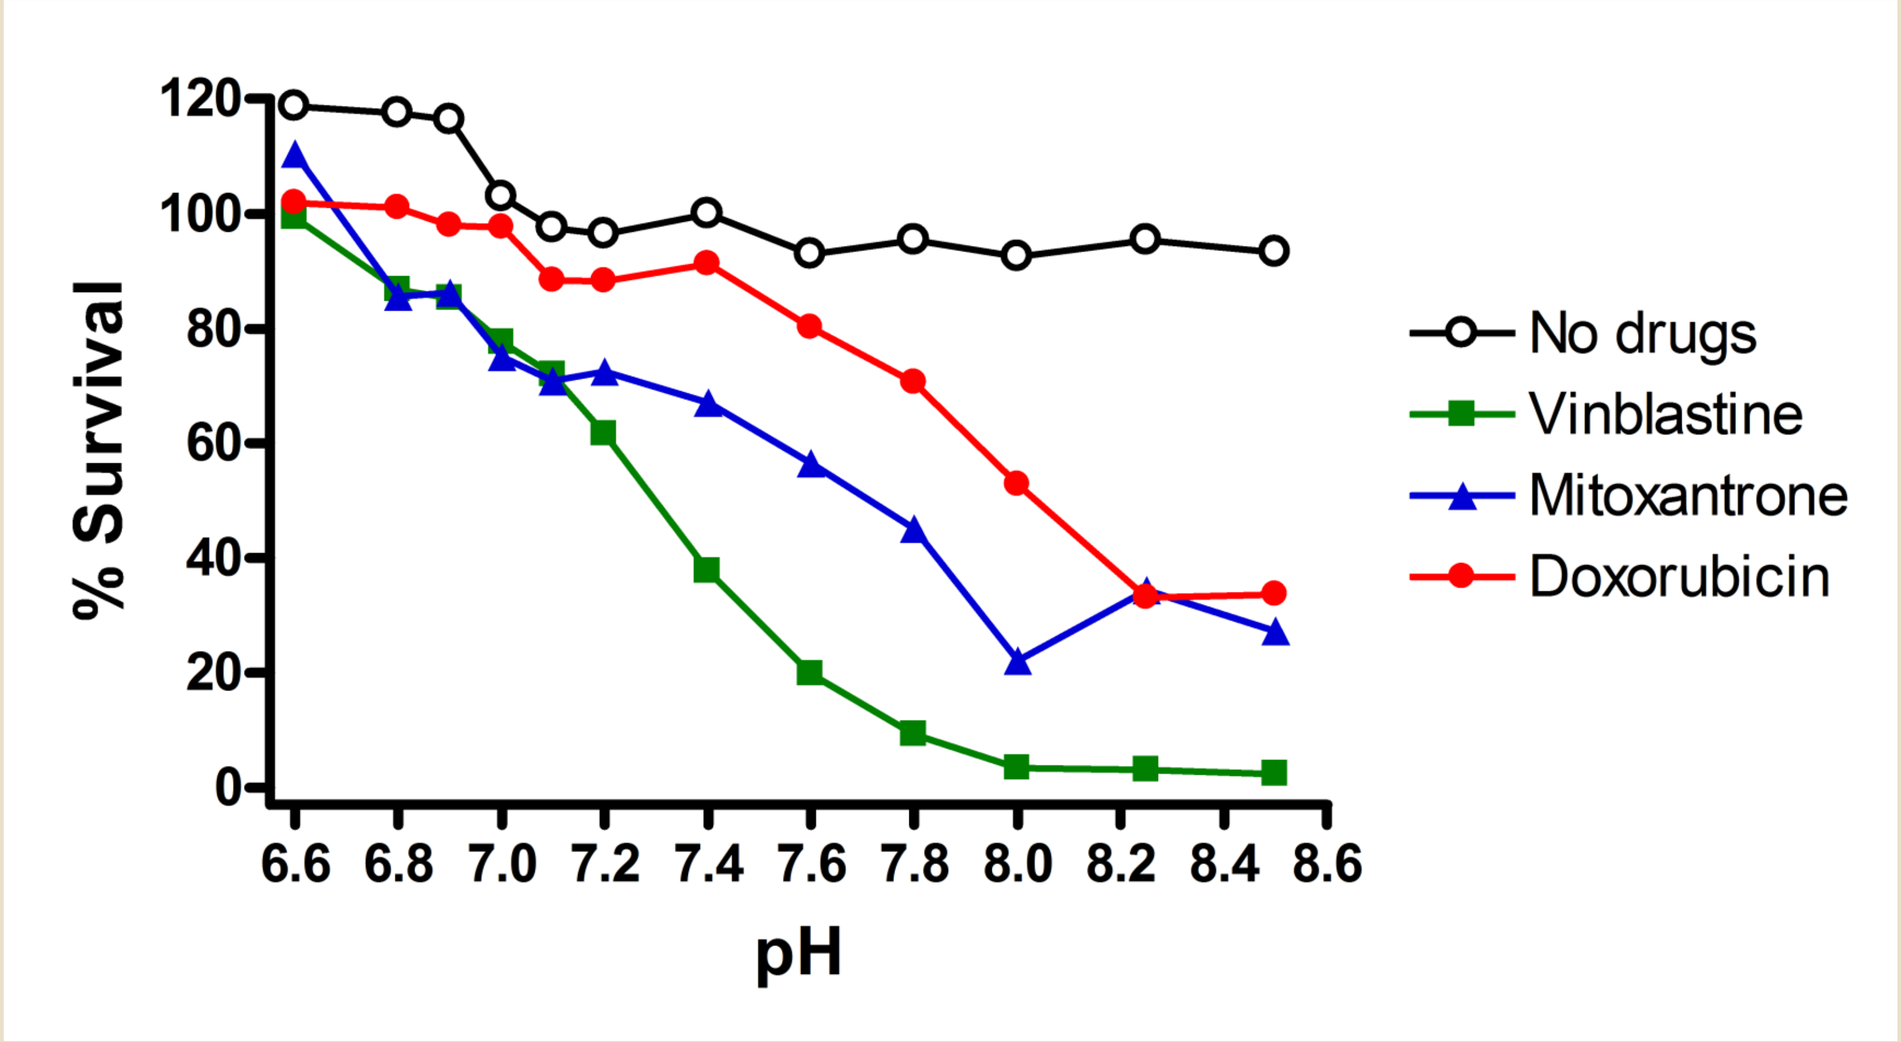
% Survival
pH
No drugs
Vinblastine
Mitoxantrone
Doxorubicin

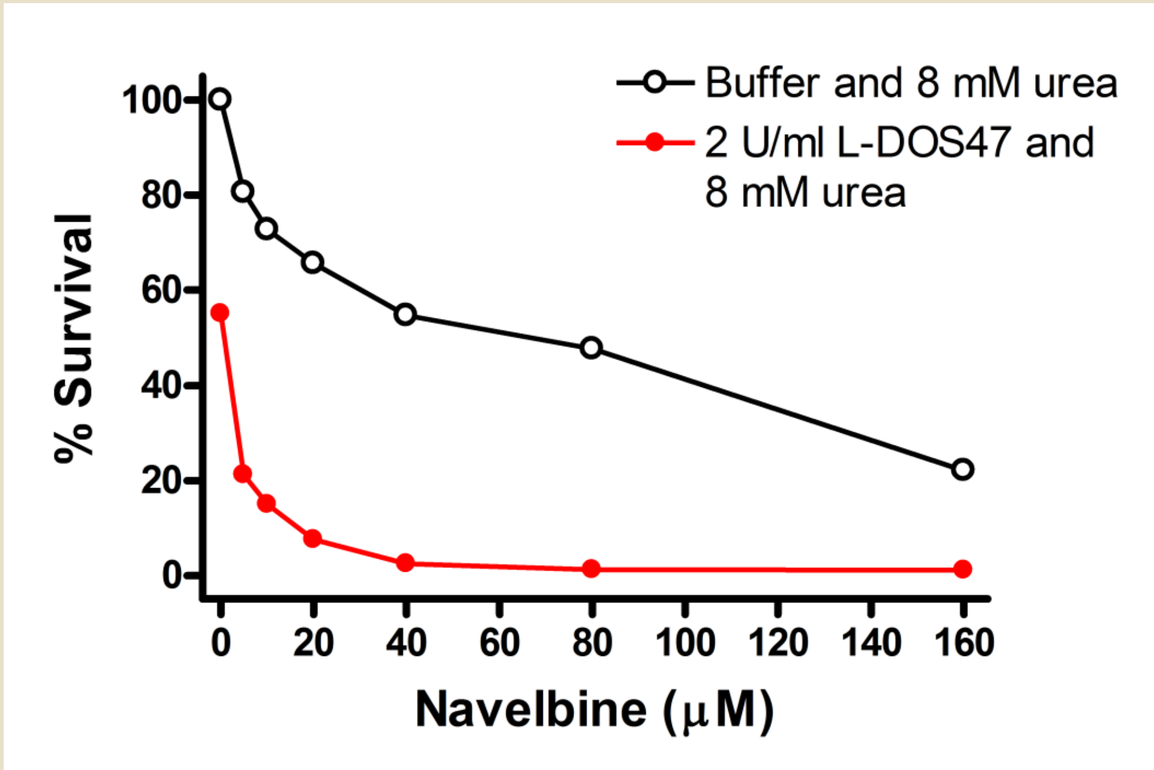
Buffer and 8 mM urea
2 U/ml L-DOS47 and 8 mM urea
% Survival
Navelbine (µM)

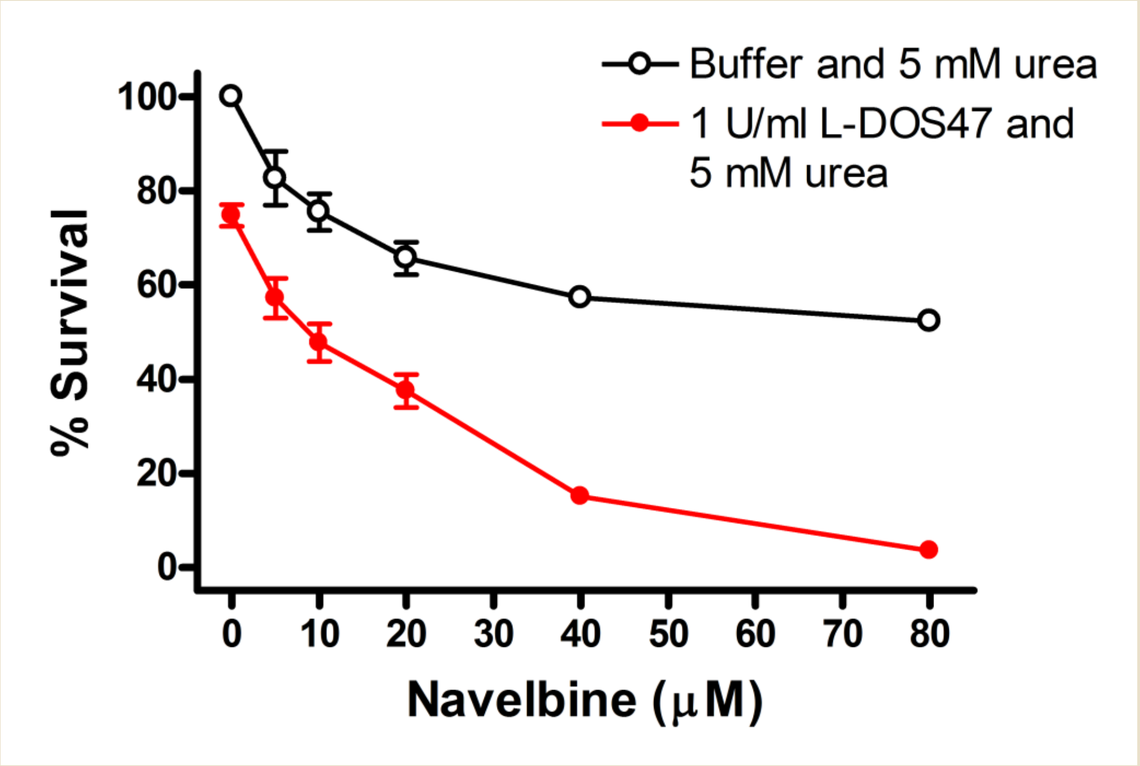
Buffer and 5 mM urea
1 U/ml L-DOS47 and 5 mM urea
% Survival
Navelbine (µM)

The cytotoxic effect of weakly basic drugs is directly related to the solution pH (top panel). At an acidic tumour pH (<6.8), the effectiveness of these drugs is significantly reduced. L-DOS47 can dramatically raise the effectiveness of these drugs (e.g. navelbine, bottom panels). This synergistic effect is directly related to its enzymatic properties of generating ammonia from urea and raising solution pH. Depending on the dosages and available urea, a 2–10 fold drug effect enhancement can be observed.

L-DOS47 binding to normal and cancer tissues

Samples	Tumor Tissue		Age-matched Normal Tissue
	Positive	Negative	Negative
Kidney carcinoma		12/12	12/12
Parathyroid adenoma		1/1	n/a
Plaenta, umbilical cord, allantois	n/a		1/1
Myofibroblastic tumor		1/1	n/a
Prostate carcinoma		4/4	4/4
Thyroid carcinoma		2/2	2/2
Pancreas adenocarcinoma	7/57 weak 8/57 v. weak	42/57	25/25
Neuroendocrine tumors		9/9	n/a
Brain, heart muscle, testis, spleen	n/a		30/30
Testis - teratoma and seminoma		3/3	3/3
Parotis tumor		1/1	1/1
Cervix squamous carcinoma		2/2	n/a
Thymoma		2/2	n/a
Colon adenocarcinoma	14/24 weak	10/24	24/24
- lymph node metastasis		3/3	
Breast adenocarcinoma		13/13	13/13
- lymph node metastasis		2/2	
Leiomoma - lung metastasis		1/1	n/a
Ovary carcinoma		4/4	n/a
Bladder carcinoma		42/42	36/36
- lymph node metastasis	1/1 strong		
- squamous carcinoma metastasis		2/2	
Lung - small cell carcinoma		1/1	5/5
- adenocarcinoma	5/5 strong		
Stomach adenocarcinoma		3/3	3/3
Liver carcinoma		4/4	4/4
Soft tissue tumors		3/3	n/a
Melanoma		48/48	18/18
- metastasis		18/18	

Immunohistochemical staining for L-DOS47 was performed on human tissue biopsies. A total of 449 tissue samples were screened. Immunopositivity is revealed by brown staining. All 5 lung adenocarcinoma and 1 bladder carcinoma metastasized to lymph node show strong positive staining pattern (over 80% of cells are positively stained). Weak positive staining (less than 5%, primarily at invasion front) is also observed in colon adenocarcinoma (14 out of 24). However, no age-matched normal tissue shows any sign of positive staining. The data are presented as number of occurrence per total number of samples in each group, where n/a denotes sample not available.

L-DOS47 binding to lung adenocarcinoma tissues

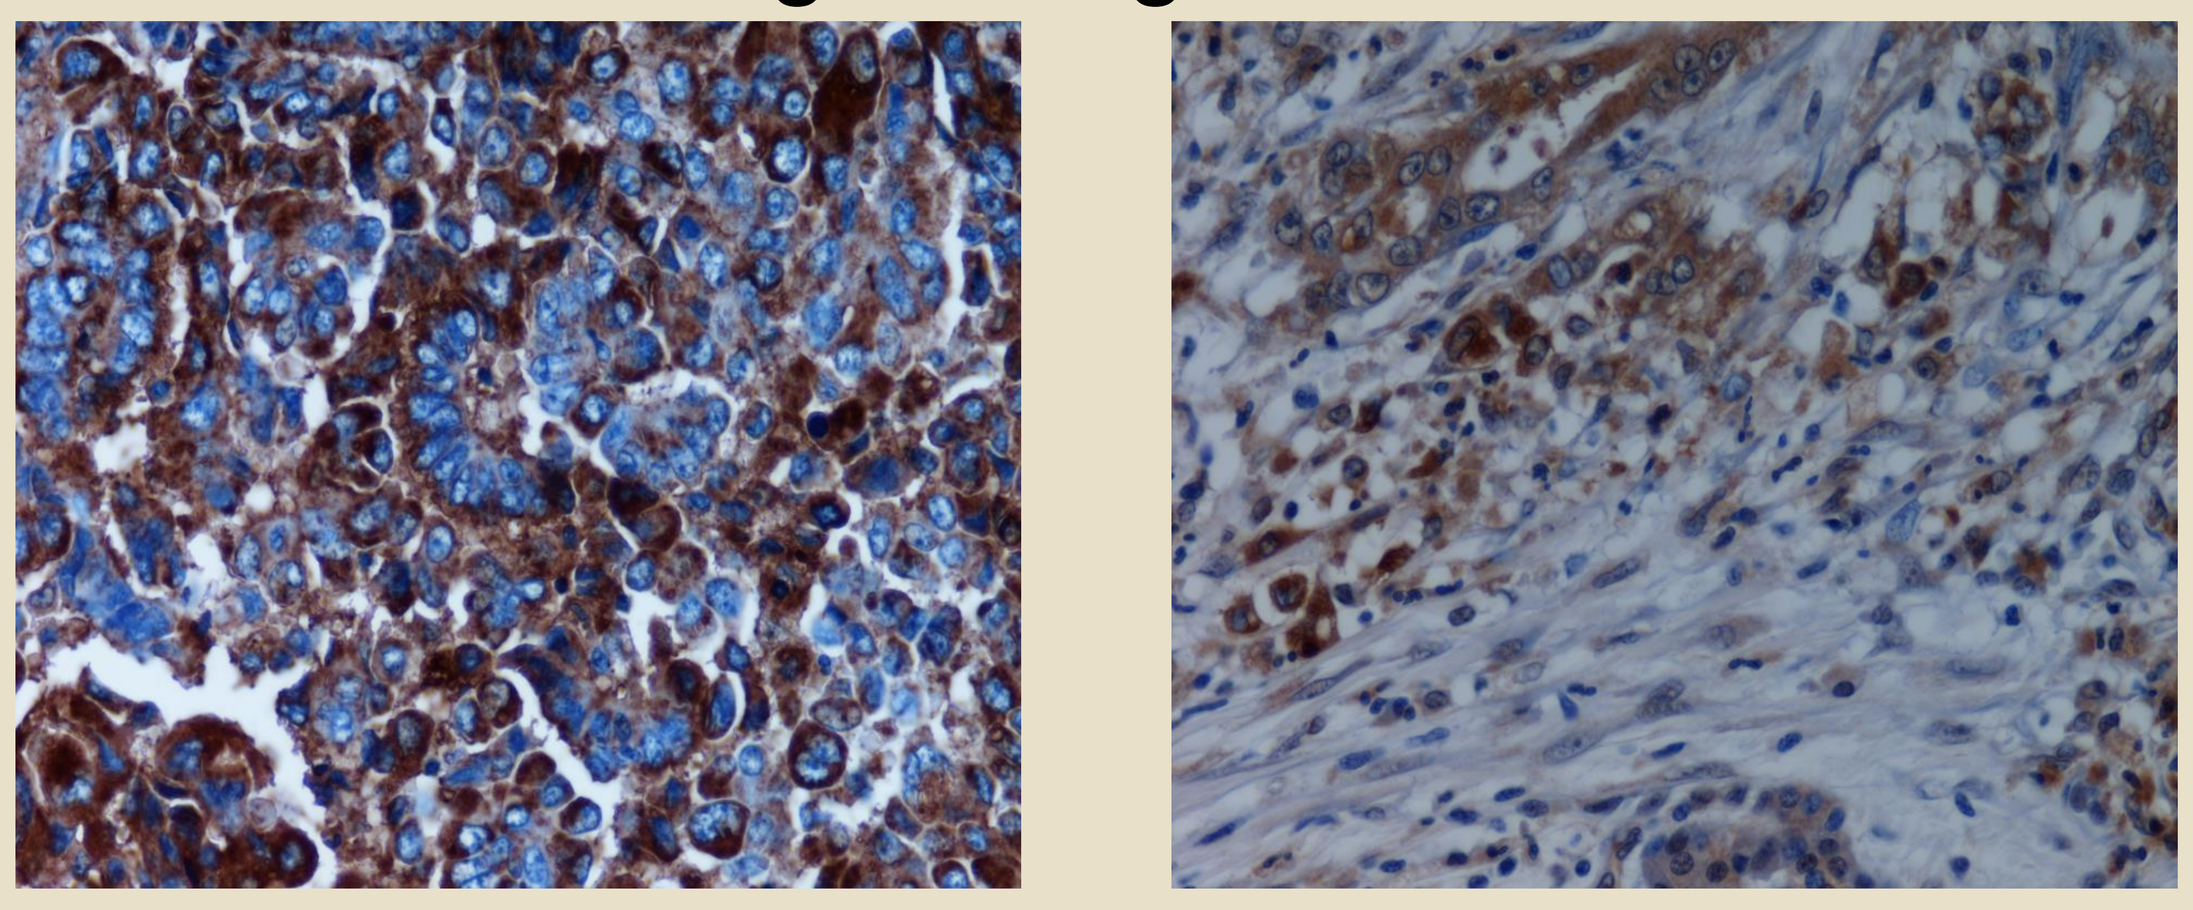

Human lung adenocarcinoma tissue biopsies were sectioned and prepared into slides. Positive binding of L-DOS47 is revealed by brown staining with blue counter stain.

L-DOS47 binding to lymph node metastases

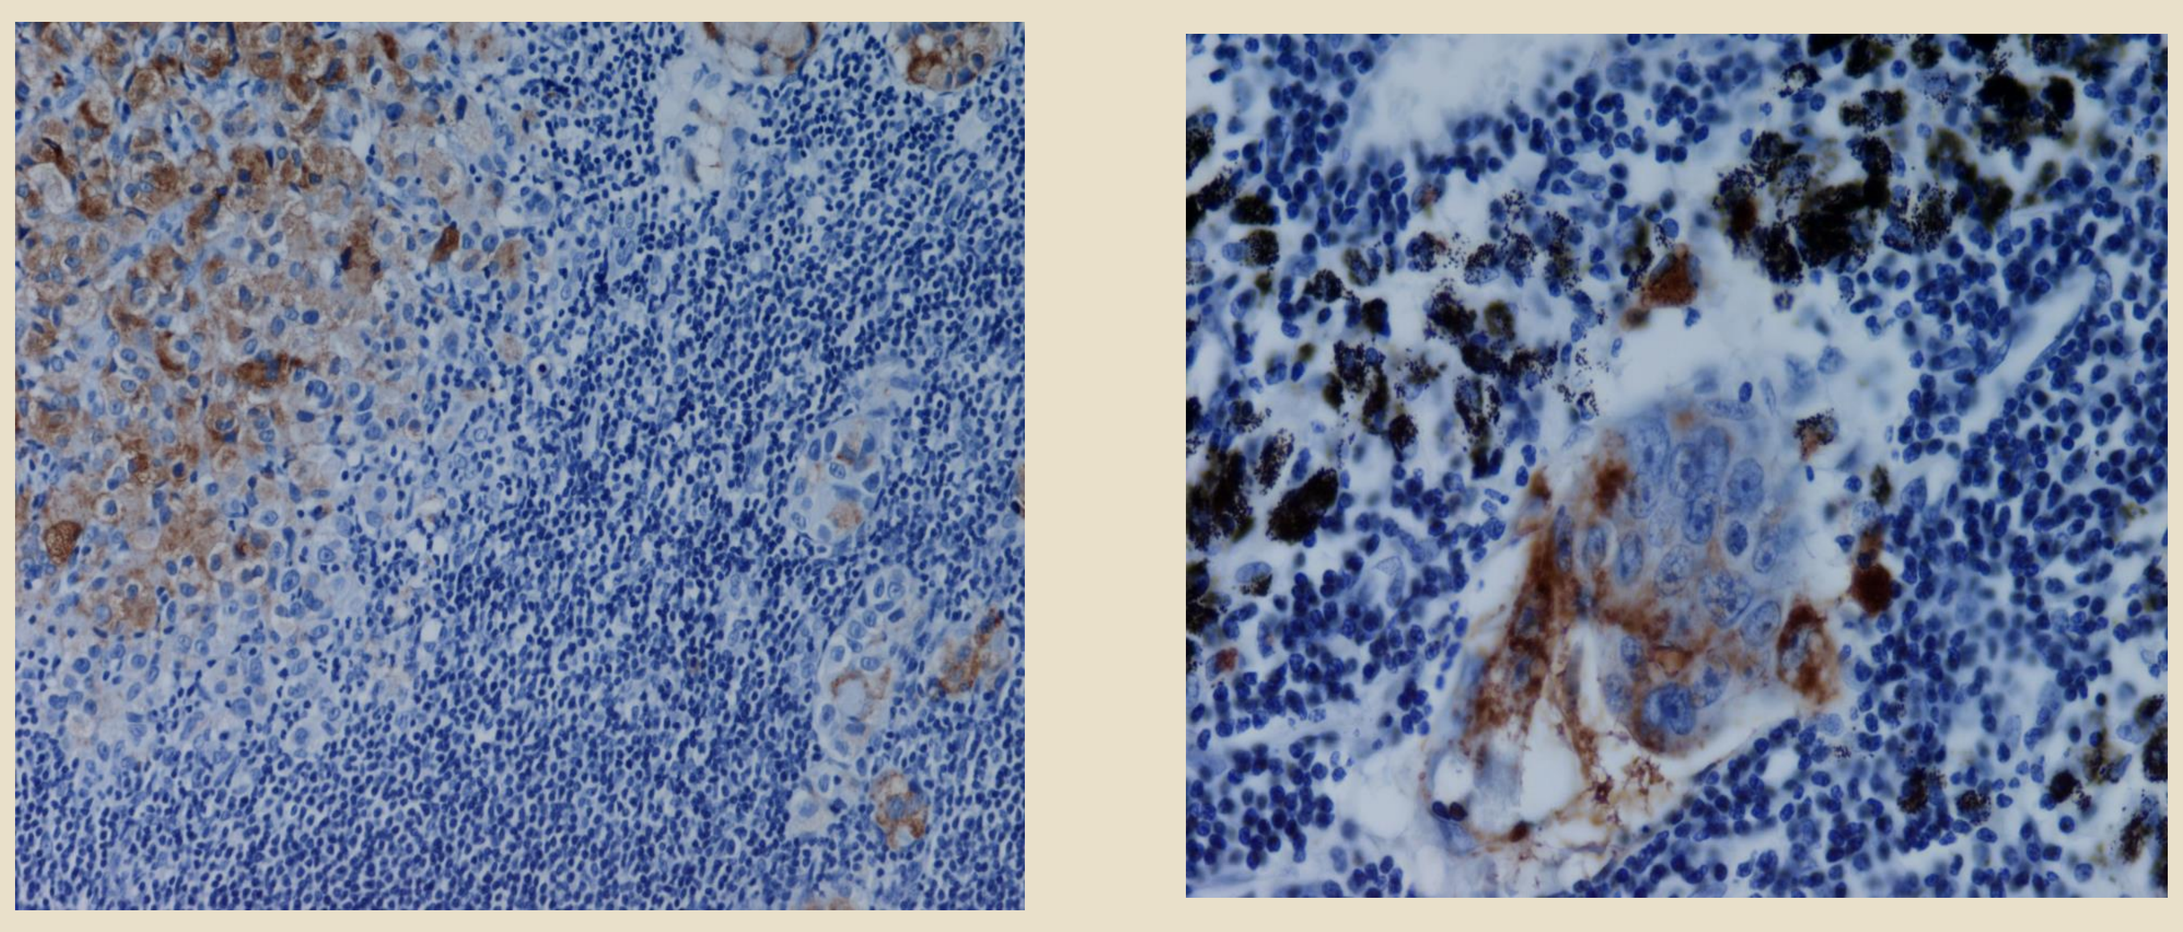

Immunopositive staining for L-DOS47 in human lung adenocarcinoma metastasized to lymph node. Positive binding of L-DOS47 is revealed by brown staining with blue counter stain. The black pigment is typical for pulmonary or mediastinal lymph node.

L-DOS47 fluorescent imaging study

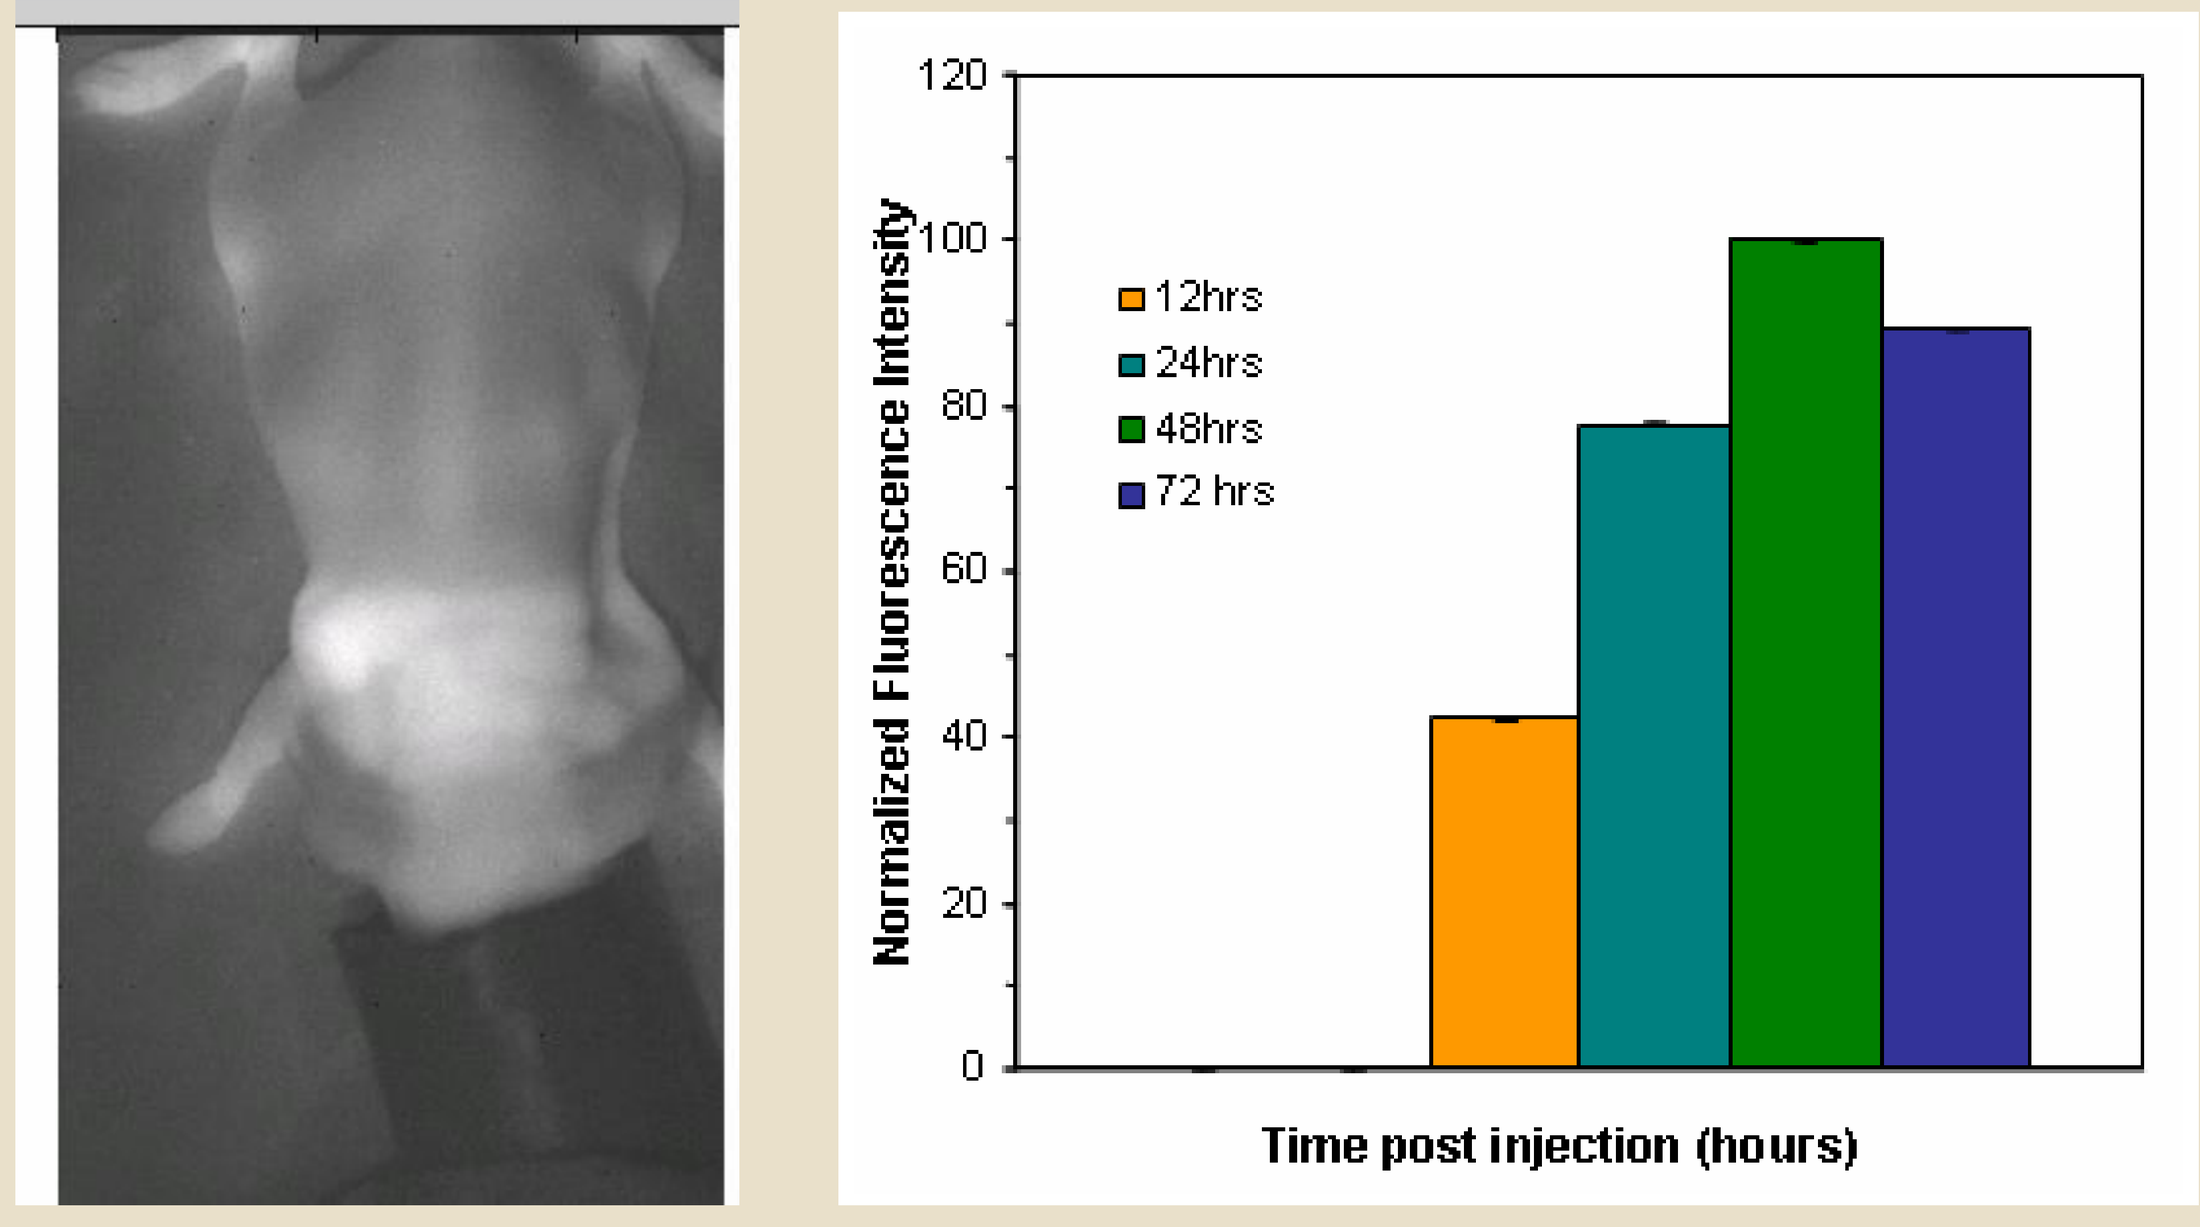
Normalized Fluorescence Intensity
12hrs
24hrs
48hrs
72 hrs
Time post injection (hours)

Inactivated L-DOS47 labelled with Cy5.5 was injected in nude mice bearing A549 xenograft. The tumour and major organs were harvested for imaging. Relative signal intensities were used to measure bio-distribution, half life and accumulation. A representative picture of a tumour bearing mouse 72h post injection of the labelled material is presented. Summary result showing signal accumulation in tumour (*ex vivo*) as a function of time is shown. Detectable L-DOS47-Cy5.5 began at 24 hours and persisted beyond 72 hours.

Cell line selection for xenograft study

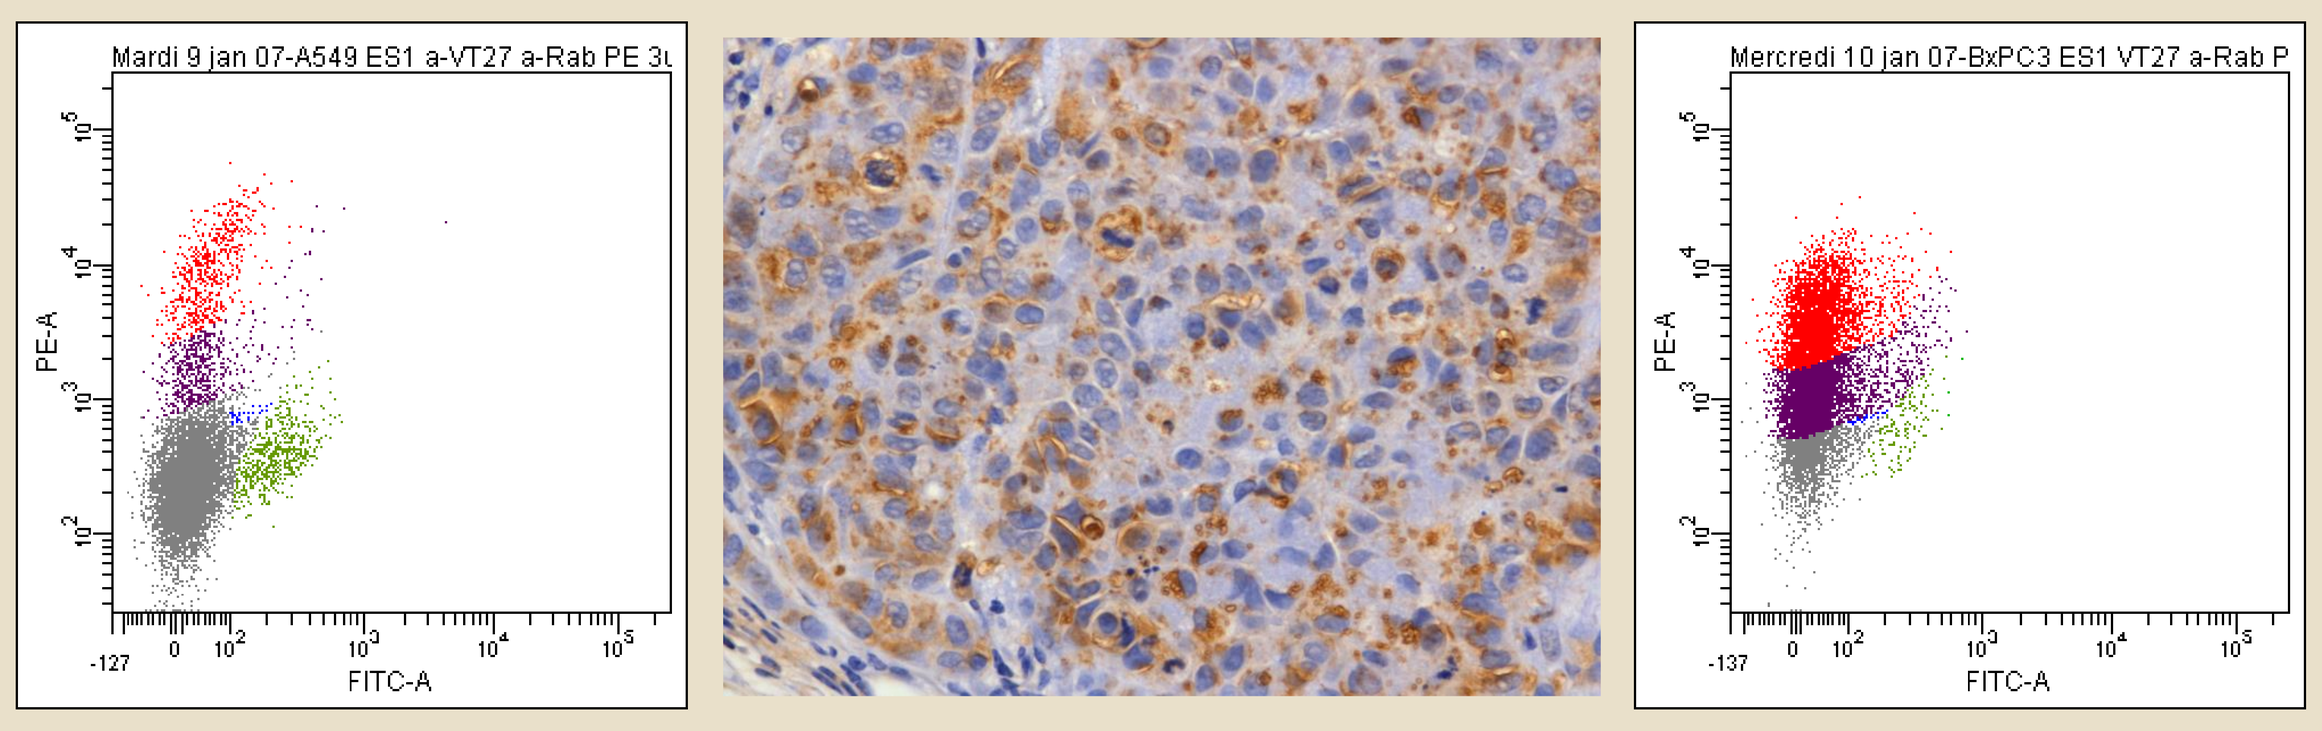
Mardi 9 jan 07-A549 ES1 a-VT27 a-Rab PE 3
PE-A
FITC-A
Mercredi 10 jan 07-BxPC3 ES1 VT27 a-Rab P
PE-A
FITC-A

Work has begun to determine the optimal xenograft model to study the drug effect of L-DOS47 *in vivo*. Previous work has shown that antigen density on commonly used lung cells such as A549 is poor (5-10%). An example of a FACS analysis of A549 cells is shown (panel left, red indicates antigen positive). A corresponding tumour slice stained with the same antibody (middle panel) revealed about 20-40% of the cells stained positively (brown stain). This suggests that A549 might not be the best model to assess efficacy. A better cell line might be BxPC-3 which over 39% of cells present the antigen (right panel, red). However, BxPC-3 is a human pancreatic adenocarcinoma line and other human pancreatic cancer tissues stained only weakly with L-DOS47. Currently the effect of L-DOS47 on both of these tumour models are being studied. The effect of antigen density on drug efficacy is also being assessed.

Conclusion

L-DOS47 is an immuno-conjugate designed to attack solid tumour by either the direct cytotoxic effect of ammonia or in combination with other weakly basic chemotherapeutic agents whose efficacies are hindered by a higher intracellular/lower extracellular pH gradient in solid tumours.

L-DOS47 is shown to have the following activities:

- Specific binding to human adenocarcinoma tissues
- Able to detect lung metastasis to lymph node
- Cytotoxic to cell lines that are recognized by the antibodies
- Significantly enhance the effect of navelbine by elevating solution pH
- Fluorescently labelled L-DOS47 accumulated specifically in A549 xenograft tumours persistently over 72h
- Studies are ongoing to determine the optimal xenograft model to use. The popular A549 model is not ideal due to the lack of antigen. A surrogate line such as BxPC-3 might be a better choice.

Future direction

Ongoing animal experiments to measure tumour pH changes *in vivo*, drug dose response and weakly basic drug potentiation effects.
Continuing preclinical studies to evaluate the toxciological and pharmacological profiles of L-DOS47 for potential human clinical studies.

Helix BioPharma Corp.

3-305 Industrial Parkway South Aurora Ontario Canada L4G 6X7 http://www.helixbiopharma.com