CAR-T cells harboring a camelid single domain antibody as a targeting agent to CEACAM6 antigen in pancreatic cancer

Wah Yau Wong[1], Jamshid Tanha[2], Lakshmi Krishnan[2], Baomin Tian[1], Praveen Kumar[1], Kim Gaspar[1], Marni D. Uger[1], Steve Demas[1], Sven Rohmann[1] and Heman Chao[1]

[1]Helix BioPharma Corp., 21 St. Clair Avenue East, Suite 1100, Toronto, Ontario M4T 1L9 Canada [2]Institute for Biological Sciences, National Research Council of Canada, Ottawa, Ontario, Canada

INTRODUCTION

Modulation of the immune system is showing tremendous promise in the treatment of malignancies. In addition to checkpoint inhibitors that re-activate T cells present in the tumor microenvironment, exogenously transduced chimeric antigen receptor (CAR) T cells are providing excellent responses in clinical trials for the treatment of leukemias. To date, the most beneficial CAR-T therapies have been directed against hematological cancers, with less success observed against solid tumors.

Carcinoembryonic Antigen Related Cell Adhesion Molecule 6 (CEACAM6) is overexpressed in many types of human cancers such as breast, pancreatic, colorectal, and non-small-cell lung adenocarcinoma, and is an independent predictor of overall survival and disease free survival. Targeting this molecule by antibodies has slowed tumor progression in certain animal models. 2A3 is a camelid single domain antibody isolated from a whole cancer cell-immunized llama library. The antibody binds specifically to the CEACAM6 antigen with high affinity (5nM as measured by surface plasmon resonance) and inhibits the proliferation of CEACAM6-expressing cancer cells *in vitro*.

In this study, we investigated the use of CAR-T cells as a therapy for pancreatic carcinoma. CAR-T cells were engineered to express the anti-CEACAM 6 antibody 2A3 in combination with the CD28 costimulatory molecule and CD3 zeta chain. Co-incubation of anti-CEACAM6 CAR-T cells with the CEACAM6-expressing pancreatic cell line BxPC3 resulted in decreased viability of the BxPC3 cells and T cell production of cytokines (IL-2 and IFN-γ), suggesting potential anti-cancer activity of the anti-CEACAM6 CAR-T cells.

The efficacy of anti-CEACAM6 CAR-T cells *in vivo* was investigated in two xenograft models using BxPC3 tumor cells. In the first study, treatment with anti-CEACAM6 CAR-T cells was initiated on the day after tumor implantation. In the second study, tumors were allowed to reach a volume of 100mm^3 before treatment was initiated. In both studies, treatment with anti-CEACAM6 CAR-T cells significantly decreased the growth of the BxPC3 tumors as compared to treatment with untransduced T cells. The results strongly support the use of anti-CEACAM6 CAR-T cells as an immunotherapeutic agent against CEACAM6-expressing solid cancers, and that camelid single domain antibodies can be easily adopted for CAR-T therapies.

ACKNOWLEDGMENTS

The authors would like to thank ProMab Biotechnologies, Inc. for performing the studies described in this poster.

Helix BioPharma Corp.
21 St. Clair Avenue East, Suite 1100
Toronto, Ontario M4T 1L9 Canada
http://www.helixbiopharma.com

RESULTS

Expression of the 2A3 llama anti-CEACAM6 antibody by CAR-T cells

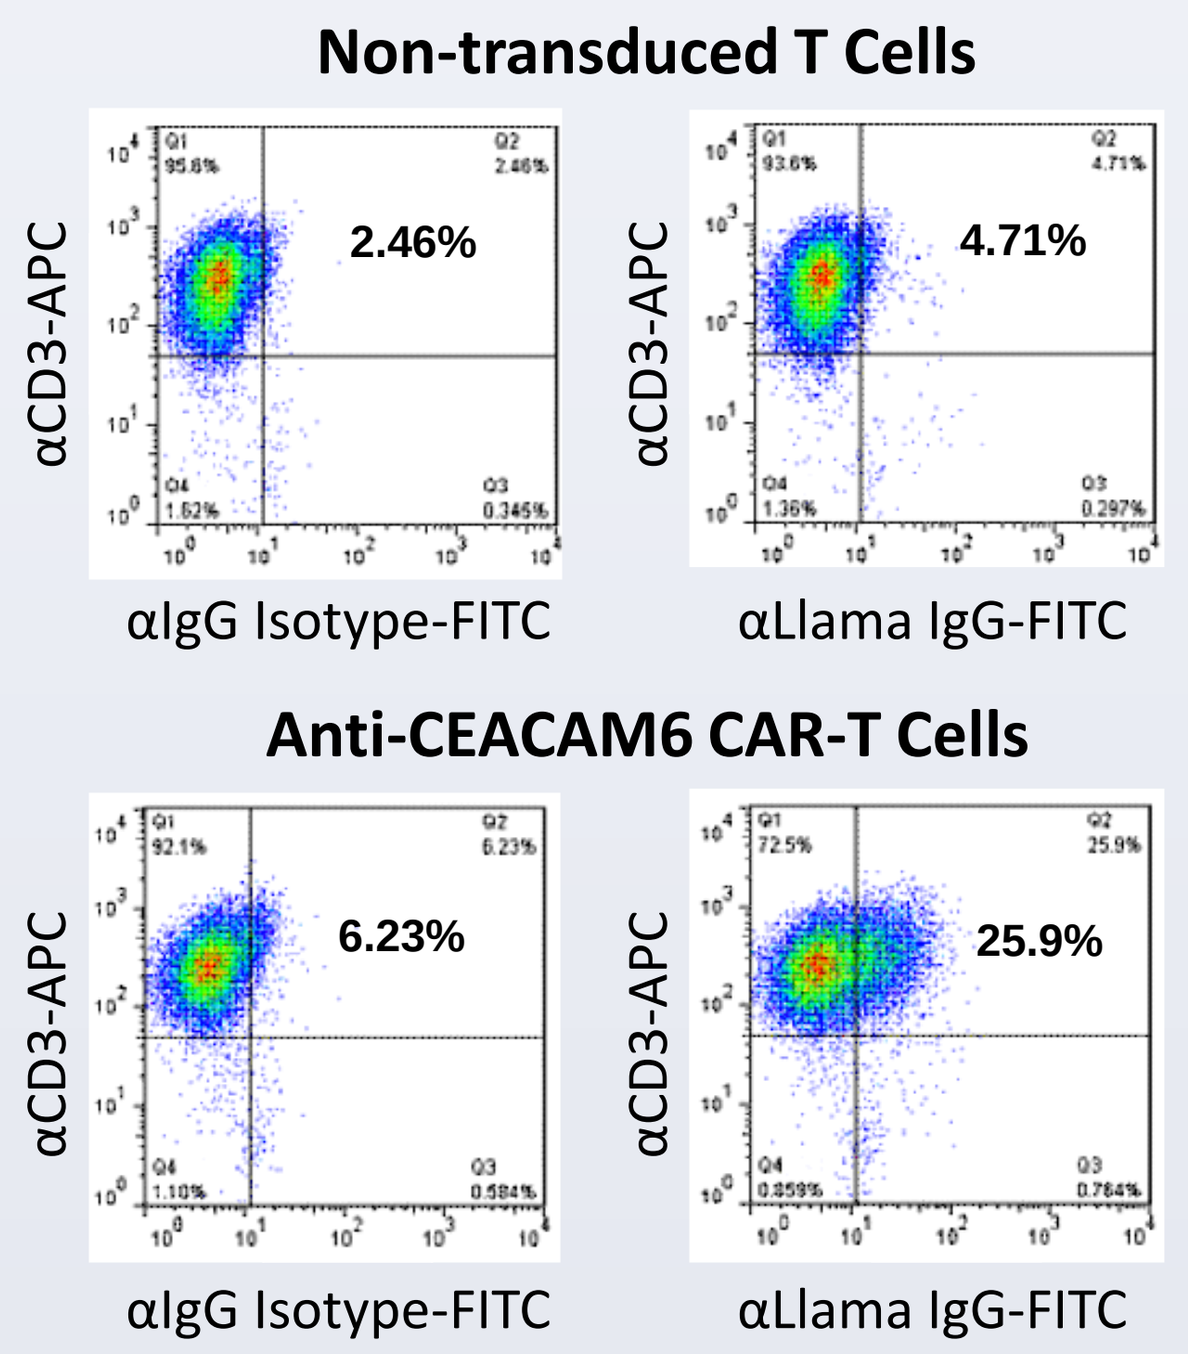
Non-transduced T Cells
αCD3-APC
2.46%
4.71%
αIgG Isotype-FITC
αLlama IgG-FITC
Anti-CEACAM6 CAR-T Cells
6.23%
25.9%
αIgG Isotype-FITC
αLlama IgG-FITC

Figure 1: PBMC were activated with CD28/CD3 microbeads and incubated with IL-2. After 24 hours, cells were transduced with a lentivirus expressing the anti-CEACAM6-CD28-CD3 construct. Flow cytometry was performed after 13 days of cell expansion. Cells were stained with anti-CD3-APC to show a high proportion of T-cells, and with rabbit anti-llama IgG (followed by anti-rabbit IgG-FITC) to evaluate the percentage of cells expressing the 2A3 anti-CEACAM6 antibody. Transduction efficiency is approximately 20%.

Cytokine production from anti-CEACAM6 CAR-T cells incubated with BxPC3 cells

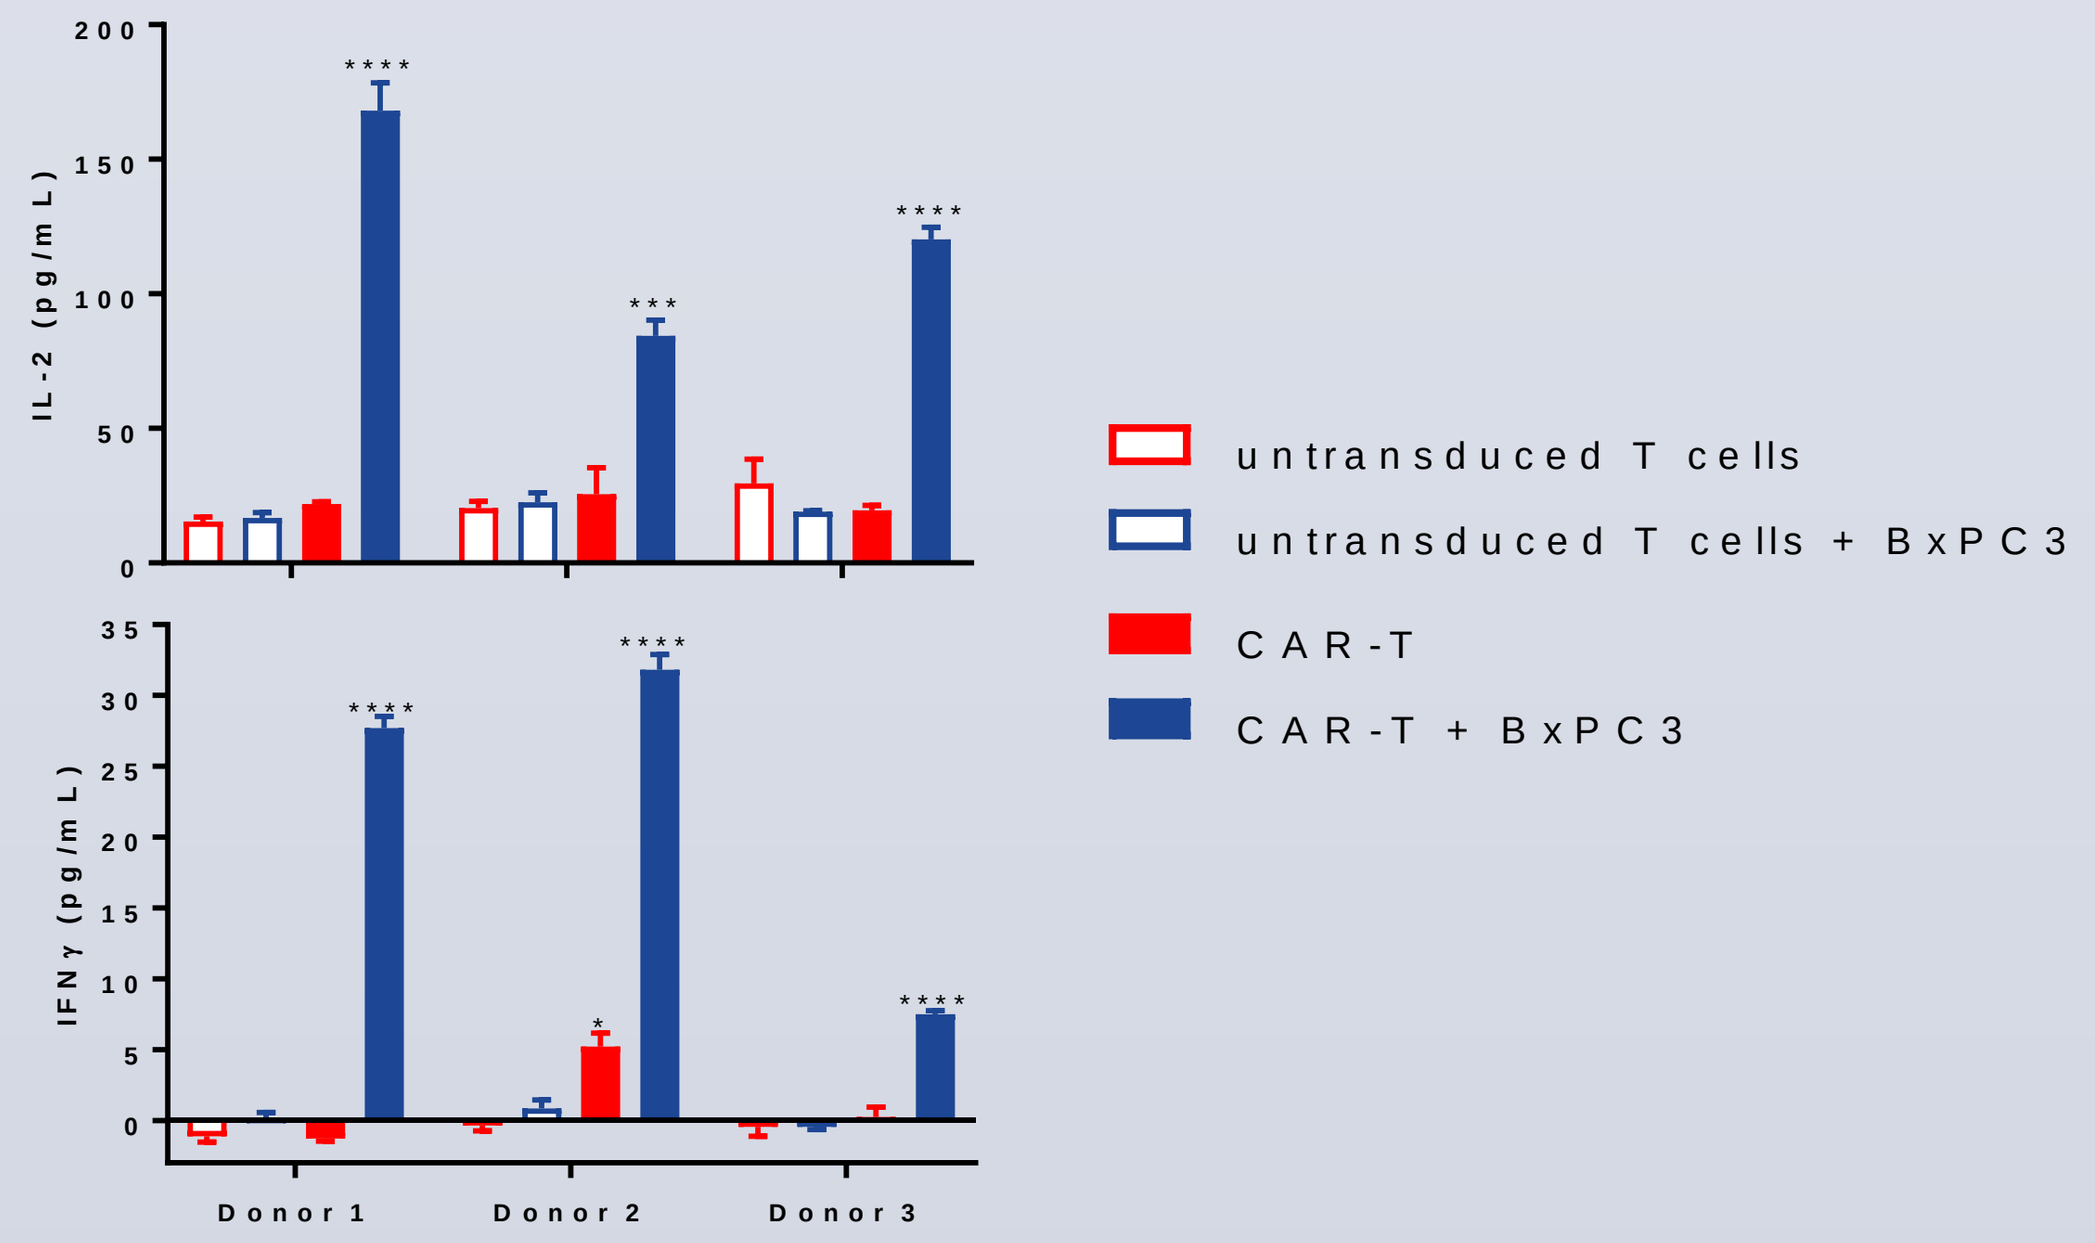
IL-2 (pg/mL)
IFNγ (pg/mL)
Donor 1
Donor 2
Donor 3
untransduced T cells
untransduced T cells + BxPC3
CAR-T
CAR-T + BxPC3

Figure 3: CAR-T were expanded for 15 days before use. BxPC3 cells were plated at 10^4 cells/well and CAR-T were added at a 1:1 E:T ratio. Cells were co-cultured for approximately 18 hours before removal of supernatant. IL-2 and IFNγ levels in the supernatant were determined by ELISA. There is a highly statistically significant difference in the amount of IL-2 and IFNγ produced by all three donor T cells when CAR-T are incubated with BxPC3 (**** p<0.0001, *** p=0.0003-0.0006, *p=0.005-0.02).

Cell viability of BxPC3 cells treated with anti-CEACAM6 CAR-T cells generated from three different donor PBMC

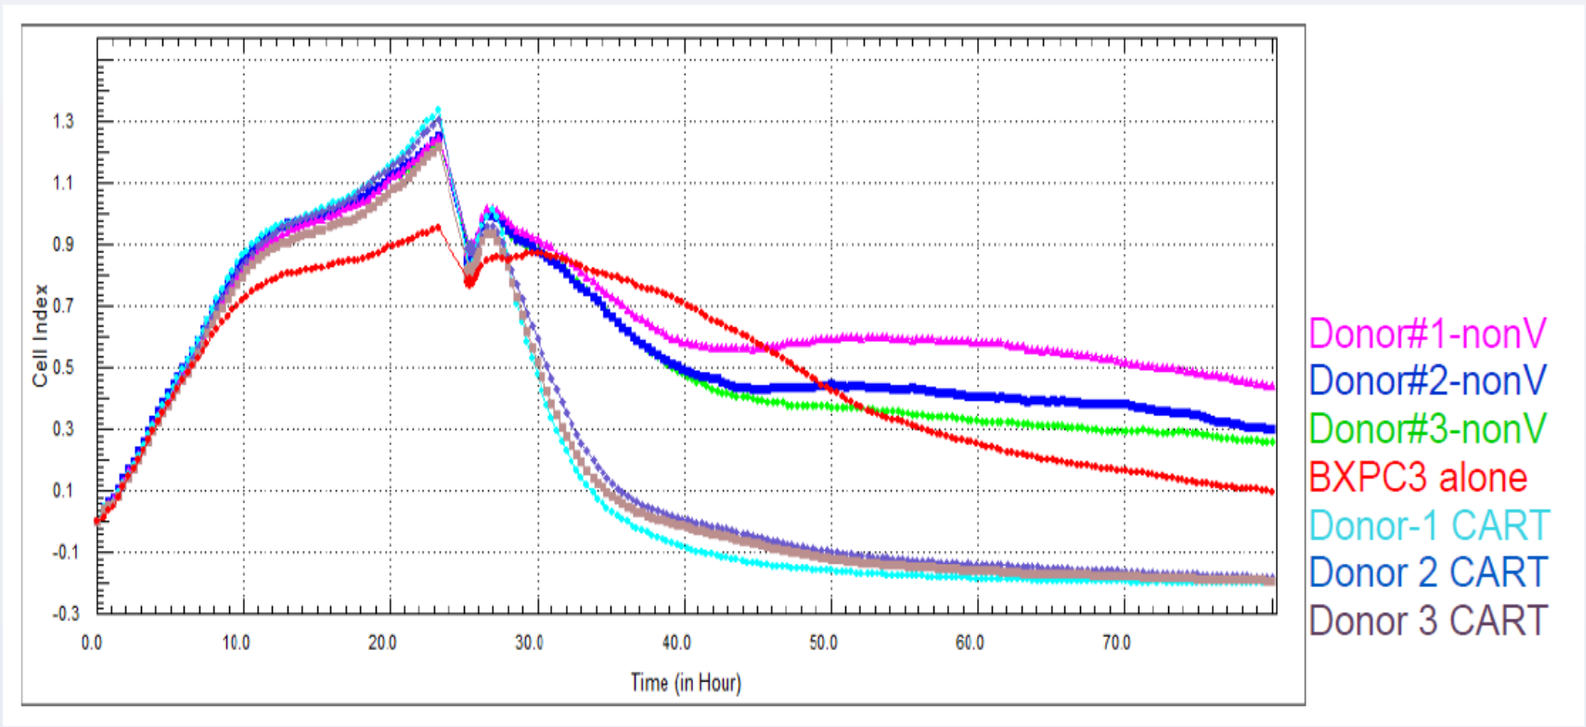
Cell Index
Time (in Hour)
Donor#1-nonV
Donor#2-nonV
Donor#3-nonV
BXPC3 alone
Donor-1 CART
Donor 2 CART
Donor 3 CART

Figure 2: CAR-T were expanded for 14-15 days before use. BxPC3 cells were plated at 10^4 cells/well and incubated for 24 hours before addition of effector CAR-T cells or untransduced T cells (nonV) at a 10:1 E:T ratio. Cell viability was evaluated using the Real Time Cell Analysis (RTCA) system from ACEA Biosciences. Anti-CEACAM6 CAR-T cells generated by all three donors decreased the viability of BxPC3 cells, whereas there was only a minimal effect on cell viability when untransduced T cells were added.

Efficacy of anti-CEACAM6 CAR-T cells in a BxPC3 preventative xenograft model

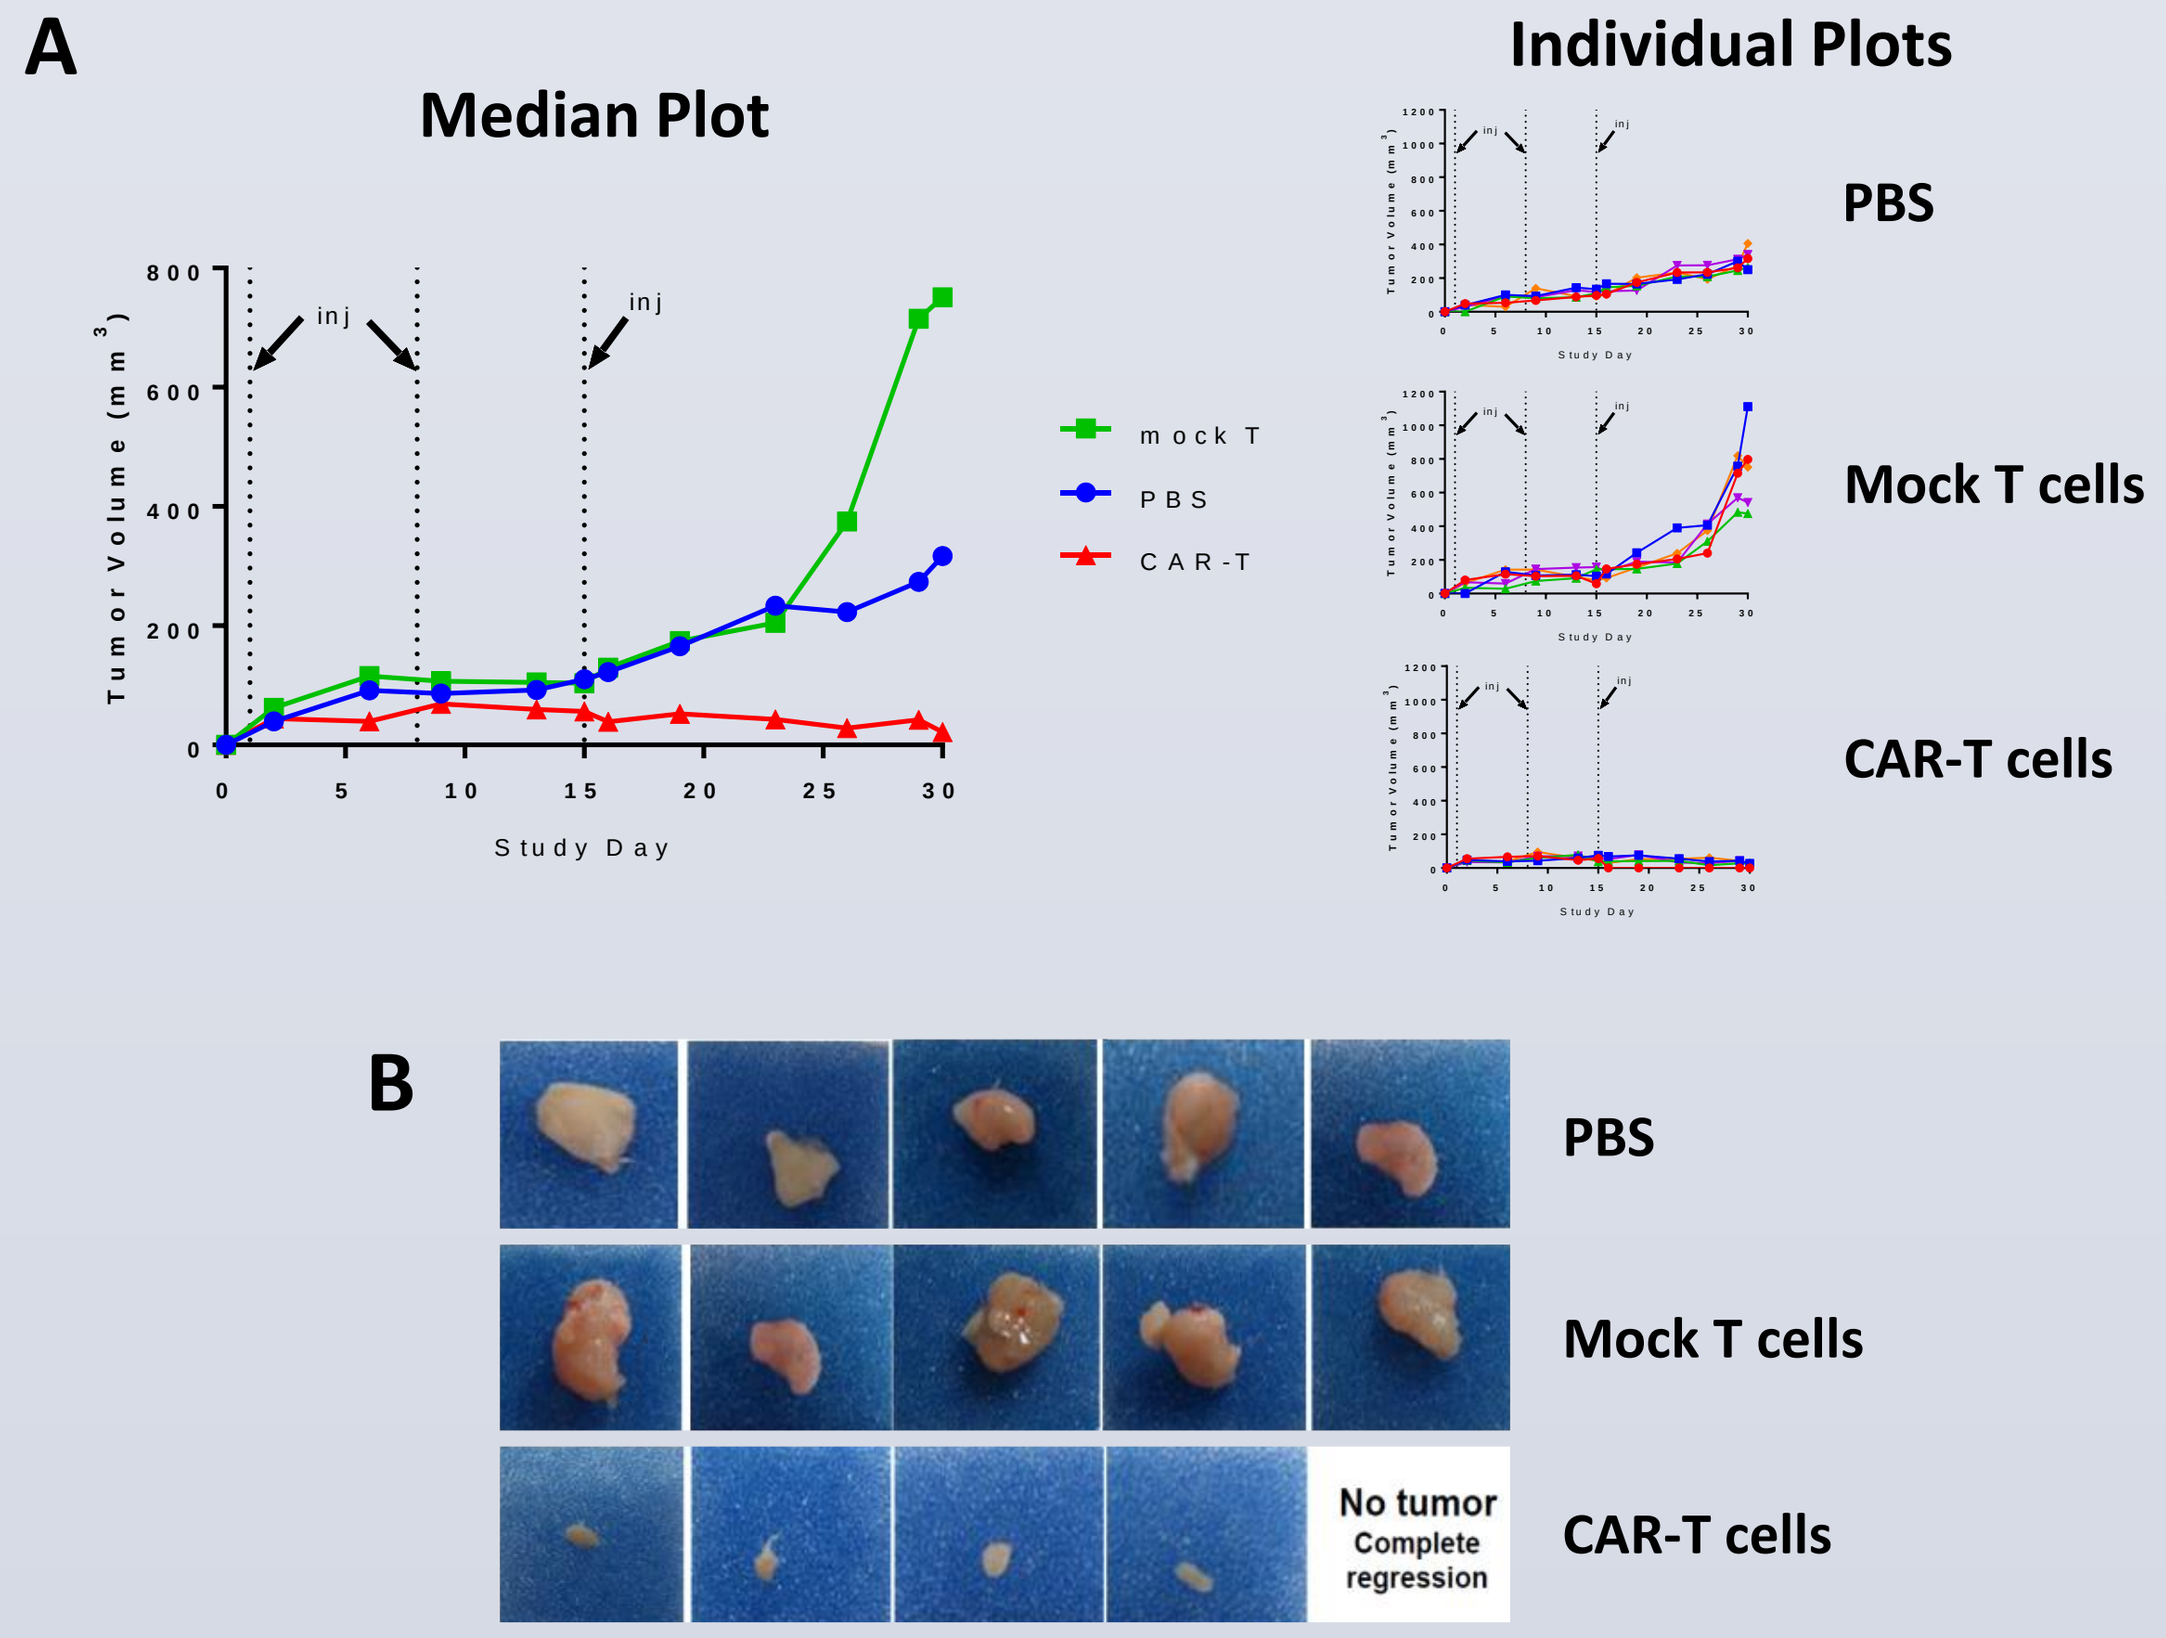
A
Individual Plots
Median Plot
Tumor Volume (mm3)
Study Day
inj
mock T
PBS
CAR-T
PBS
Mock T cells
CAR-T cells
B
PBS
Mock T cells
No tumor
Complete regression
CAR-T cells

Figure 4: BxPC3 cells (2x10^6 cells/mouse) were injected subcutaneously into the hind flank of CIEA NOG female mice. Mice were treated with either PBS, untransduced T cells (mock T cells) or anti-CEACAM6 CAR-T cells (10^7 cells/mouse) intravenously into the tail vein on days 1, 8 and 15 after tumor injections. (A) Tumor size was measured with calipers, and tumor volume was calculated using the formula (length x width^2)/2. Treatment with anti-CEACAM6 CAR-T cells significantly decreased the growth of the BxPC3 xenografts (ANOVA performed on day 30 data shows p=0.0212 for PBS vs CAR-T cells and p<0.0001 for mock T cells vs CAR-T cells). (B) Photographs of tumors at the end of the study.

Efficacy of anti-CEACAM6 CAR-T cells in a BxPC3 treatment xenograft model

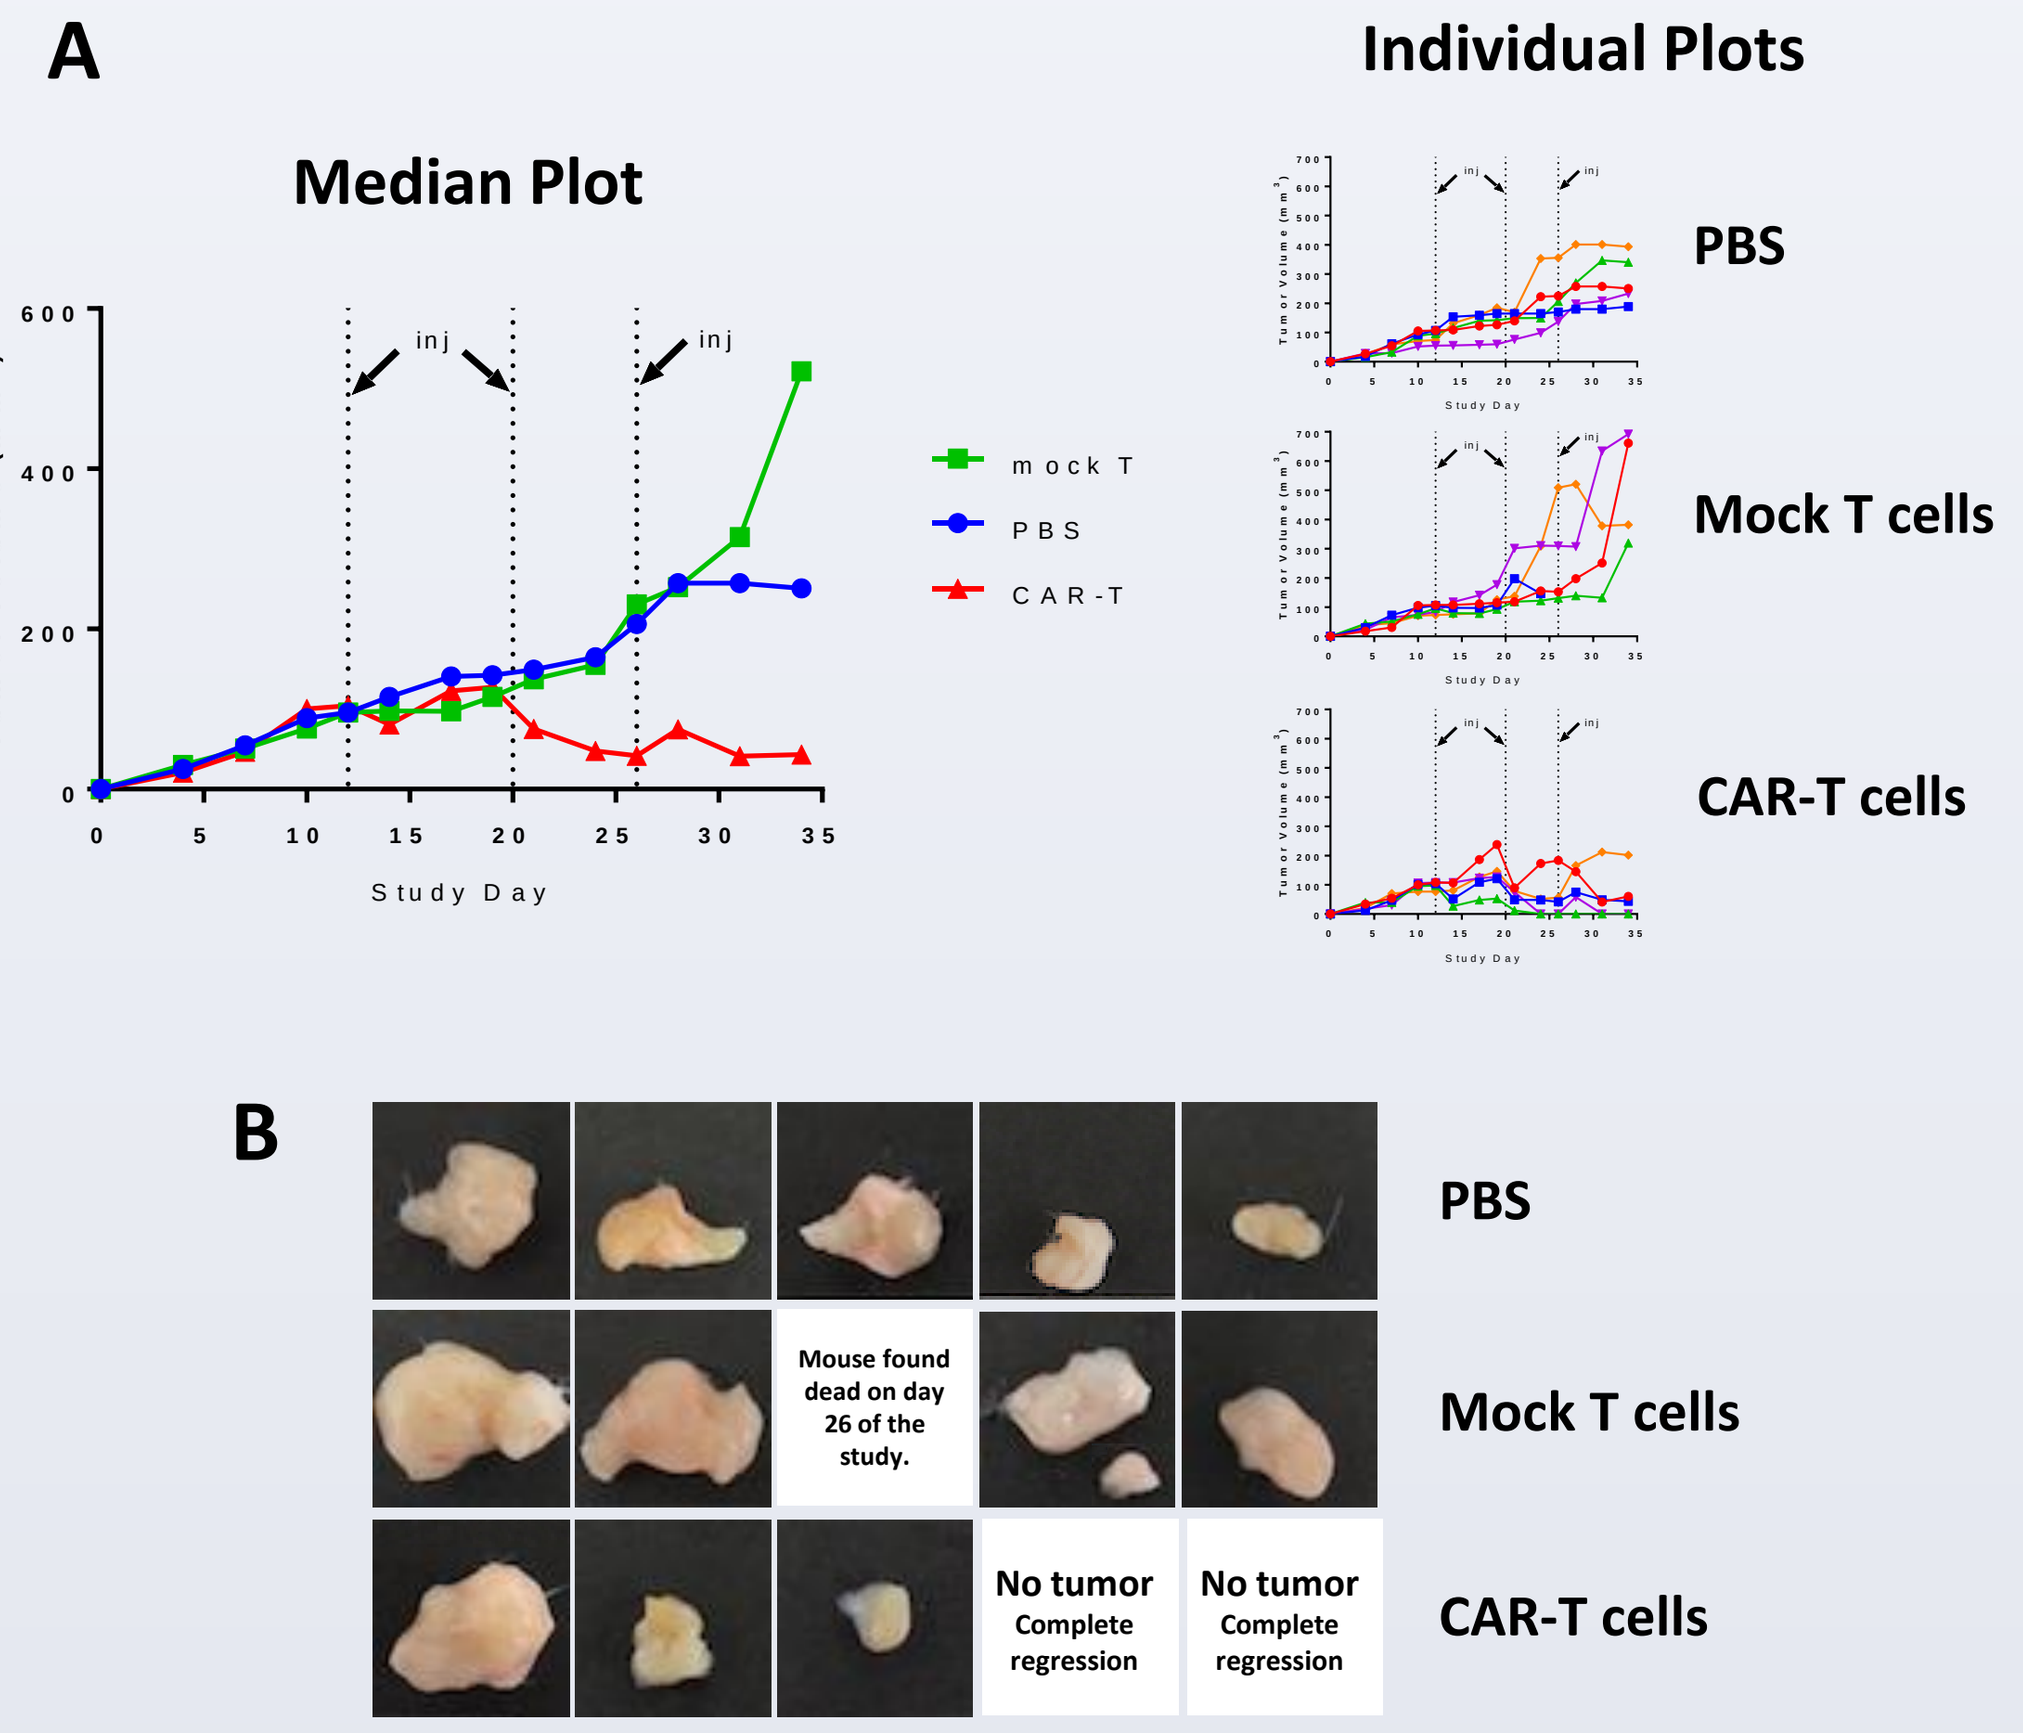
A
Individual Plots
Median Plot
Tumor Volume (mm3)
Study Day
inj
mock T
PBS
CAR-T
PBS
Mock T cells
CAR-T cells
B
PBS
Mouse found dead on day 26 of the study.
Mock T cells
No tumor
Complete regression
No tumor
Complete regression
CAR-T cells

Figure 5: BxPC3 cells were injected subcutaneously into the hind flank of CIEA NOG female mice. Mice were treated with either PBS, untransduced T cells (mock T cells) or anti-CEACAM6 CAR-T cells intravenously into the tail vein on days 12, 20 and 26 after tumor injections. (A) Tumor size was measured with calipers, and tumor volume was calculated using the formula (length x width^2)/2. Treatment with anti-CEACAM6 CAR-T cells significantly decreased the growth of the BxPC3 xenografts (ANOVA performed on day 34 data shows p=0.0386 for PBS vs CAR-T cells and p=0.0005 for mock T cells vs CAR-T cells). (B) Photographs of tumors at the end of the study.

CONCLUSIONS

- CAR-T cells generated against the CEACAM6 antigen are highly effective at reducing the cell viability of the CEACAM6-expressing pancreatic cancer cell line BxPC3 *in vitro*
- CAR-T cells generated against the CEACAM6 antigen significantly reduce the growth of the BxPC3 pancreatic carcinoma *in vivo* both when used in a preventative tumor model and in a model where treatment is initiated after the tumor is established
- Camelid single chain antibodies can be easily adopted for use in CAR-T therapies

REFERENCES

Baral, T.N., Y. Murad, T. Nguyen, U. Iqbal and J. Zhang. Isolation of functional single domain antibody by whole cell immunization: Implications for cancer treatment. 2011; 371:70-80.

Cheng, T., Y.M. Murad, C. Chang, M. Yang, T.N. Baral, A. Cowan, S. Tseng, A. Wong, R. MacKenzie, D. Shieh and J. Zhang. Single domain antibody against carcinoembryonic antigen-related cell adhesion molecule 6 (CEACAM6) inhibits proliferation, migration, invasion and angiogenesis of pancreatic cancer cells. 2014; 50:713-721.

RESEARCH POSTER PRESENTATION DESIGN © 2015
www.PosterPresentations.com